PLAG (1-Palmitoyl-2-Linoleoyl-3-Acetyl-rac-Glycerol) modulates eosinophil chemotaxis by regulating CCL26 expression from epithelial cells

Jinseon Jeong[1,2], Young-Jun Kim[1], Sun Young Yoon[3], Young-Jae Kim[1], Joo Heon Kim[4], Myung-Hwan Kim[5], Ki-Young Sohn[3], Heung-Jae Kim[3], Yong-Hae Han[3], Saeho Chong[3], Jae Wha Kim[1,2]

[1]Biomedical Translational Research Center, KRIBB, Daejeon 305-806, Republic of Korea. [2] Department of Functional Genomics, University of Science & Technology, Daejeon 34113, Republic of Korea. [3]Enzychem Lifesciences Corporation, Fort Lee, NJ 07024, USA. [4]Department of Pathology, Eulji University School of Medicine, Daejeon 302-120, Republic of Korea. [5]Department of Gastroenterology, Department of Oncology, Asan Medical Center, University of Ulsan College of Medicine, Seoul 138-736, Republic of Korea.

825

Abstract

Increased number of eosinophils in the circulation and sputum is associated with the severity of asthma. The respiratory epithelium produces chemokine (C-C motif) ligands (CCL) which recruits and activates eosinophils. A chemically synthesized monoacetyl-diglyceride, PLAG (1-palmitoyl-2-linoleoyl-3-acetyl-rac-glycerol) is a major constituent in the antlers of Sika deer (*Cervus nippon* Temminck) which has been used in oriental medicine. This study was aimed to investigate the molecular mechanism of PLAG effect on the alleviation of asthma phenotypes. A549, a human alveolar basal epithelial cell, and HaCaT, a human keratinocyte, were activated by the treatment of interleukin-4 (IL-4), and the expression of chemokines, known to be effective on the induction of eosinophil migration was analyzed by RT-PCR. The expression of IL-4 induced genes was modulated by the co-treatment of PLAG. Especially, CCL26 expression from the stimulated epithelial cells was significantly blocked by PLAG, which was confirmed by ELISA. The transcriptional activity of signal transducer and activator of transcription 6 (STAT6), activated by IL-4 mediated phosphorylation and nuclear translocation, was down-regulated by PLAG in a concentration-dependent manner. In ovalbumin-induced mouse model, the infiltration of immune cells into the respiratory tract was decreased by PLAG administration. Cytological analysis of the isolated bronchoalveolar lavage fluid (BALF) cells proved the infiltration of eosinophils was significantly reduced by PLAG. In addition, PLAG inhibited the migration of murine bone marrow-derived eosinophils, and human eosinophil cell line, EoL-1, which was induced by the addition of A549 culture medium.

Introduction

- Asthma is a chronic inflammatory airway disease in which mast cells, T cells, and eosinophils are infiltrated into the respiratory region [1].
- Eosinophils are circulating granulocytes responsible for parasite infection, allergy, and asthma [2].
- CCL26, also called eotaxin-3, is an eotaxin-family of chemokines and the most potent eosinophil chemotactic factor secreted by airway epithelial cells [3].
- CCL26 is produced in response to IL-4 via STAT6 pathway [4].
- PLAG is a synthetic monoacetyl-diglyceride, which is an effective regulator of TH2 immunity and rheumatoid arthritis [5].

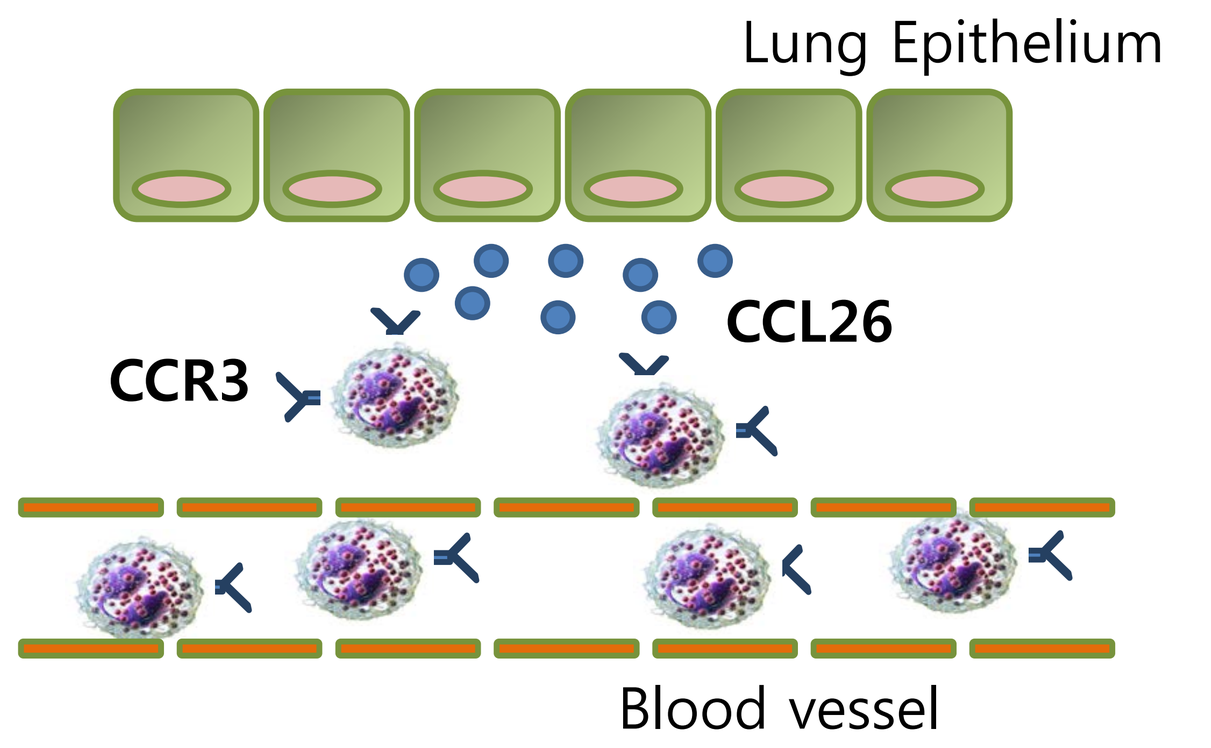

Lung Epithelium
CCL26
CCR3
Blood vessel

PLAG

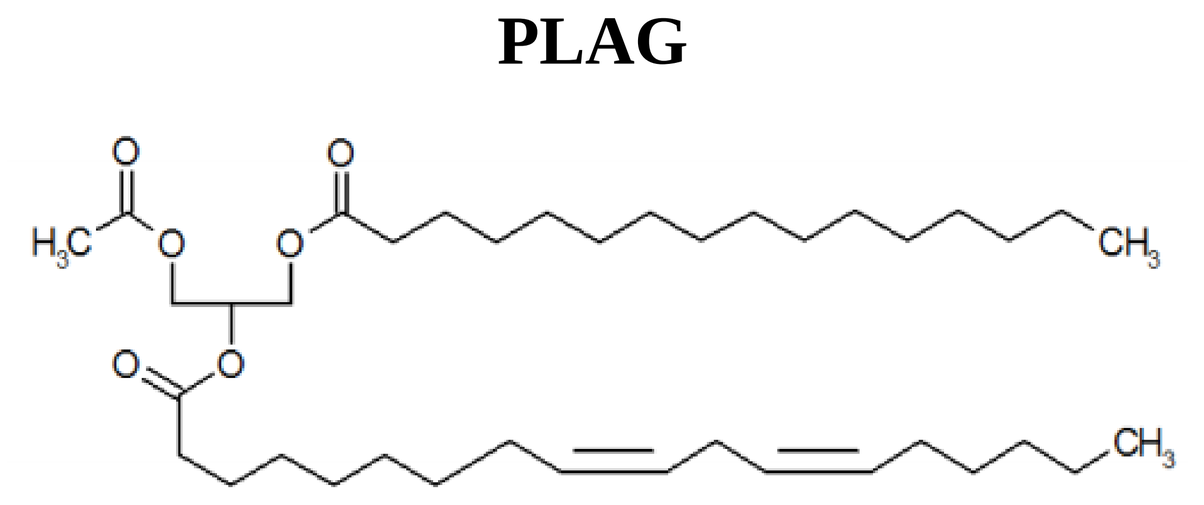

Results

1. IL-4 induced CCL26 expression is inhibited by PLAG.

We analyzed the expression of relevant genes with the regulation of eosinophil migration to find the relationship of PLAG function in asthmatic pathogenesis by RT-PCR. We found that the expression of CCL26 was significantly inhibited by PLAG in a dose-dependent manner. The effect of PLAG on IL-4 induced CCL26 secretion in A549 cells was also verified by ELISA. PLAG caused a concentration- and time-dependent inhibition of IL-4 induced CCL26 production from the epithelial cells, A549 and HaCaT. These findings suggest that PLAG is able to inhibit IL-4 induced production of CCL26 from epithelial cells.

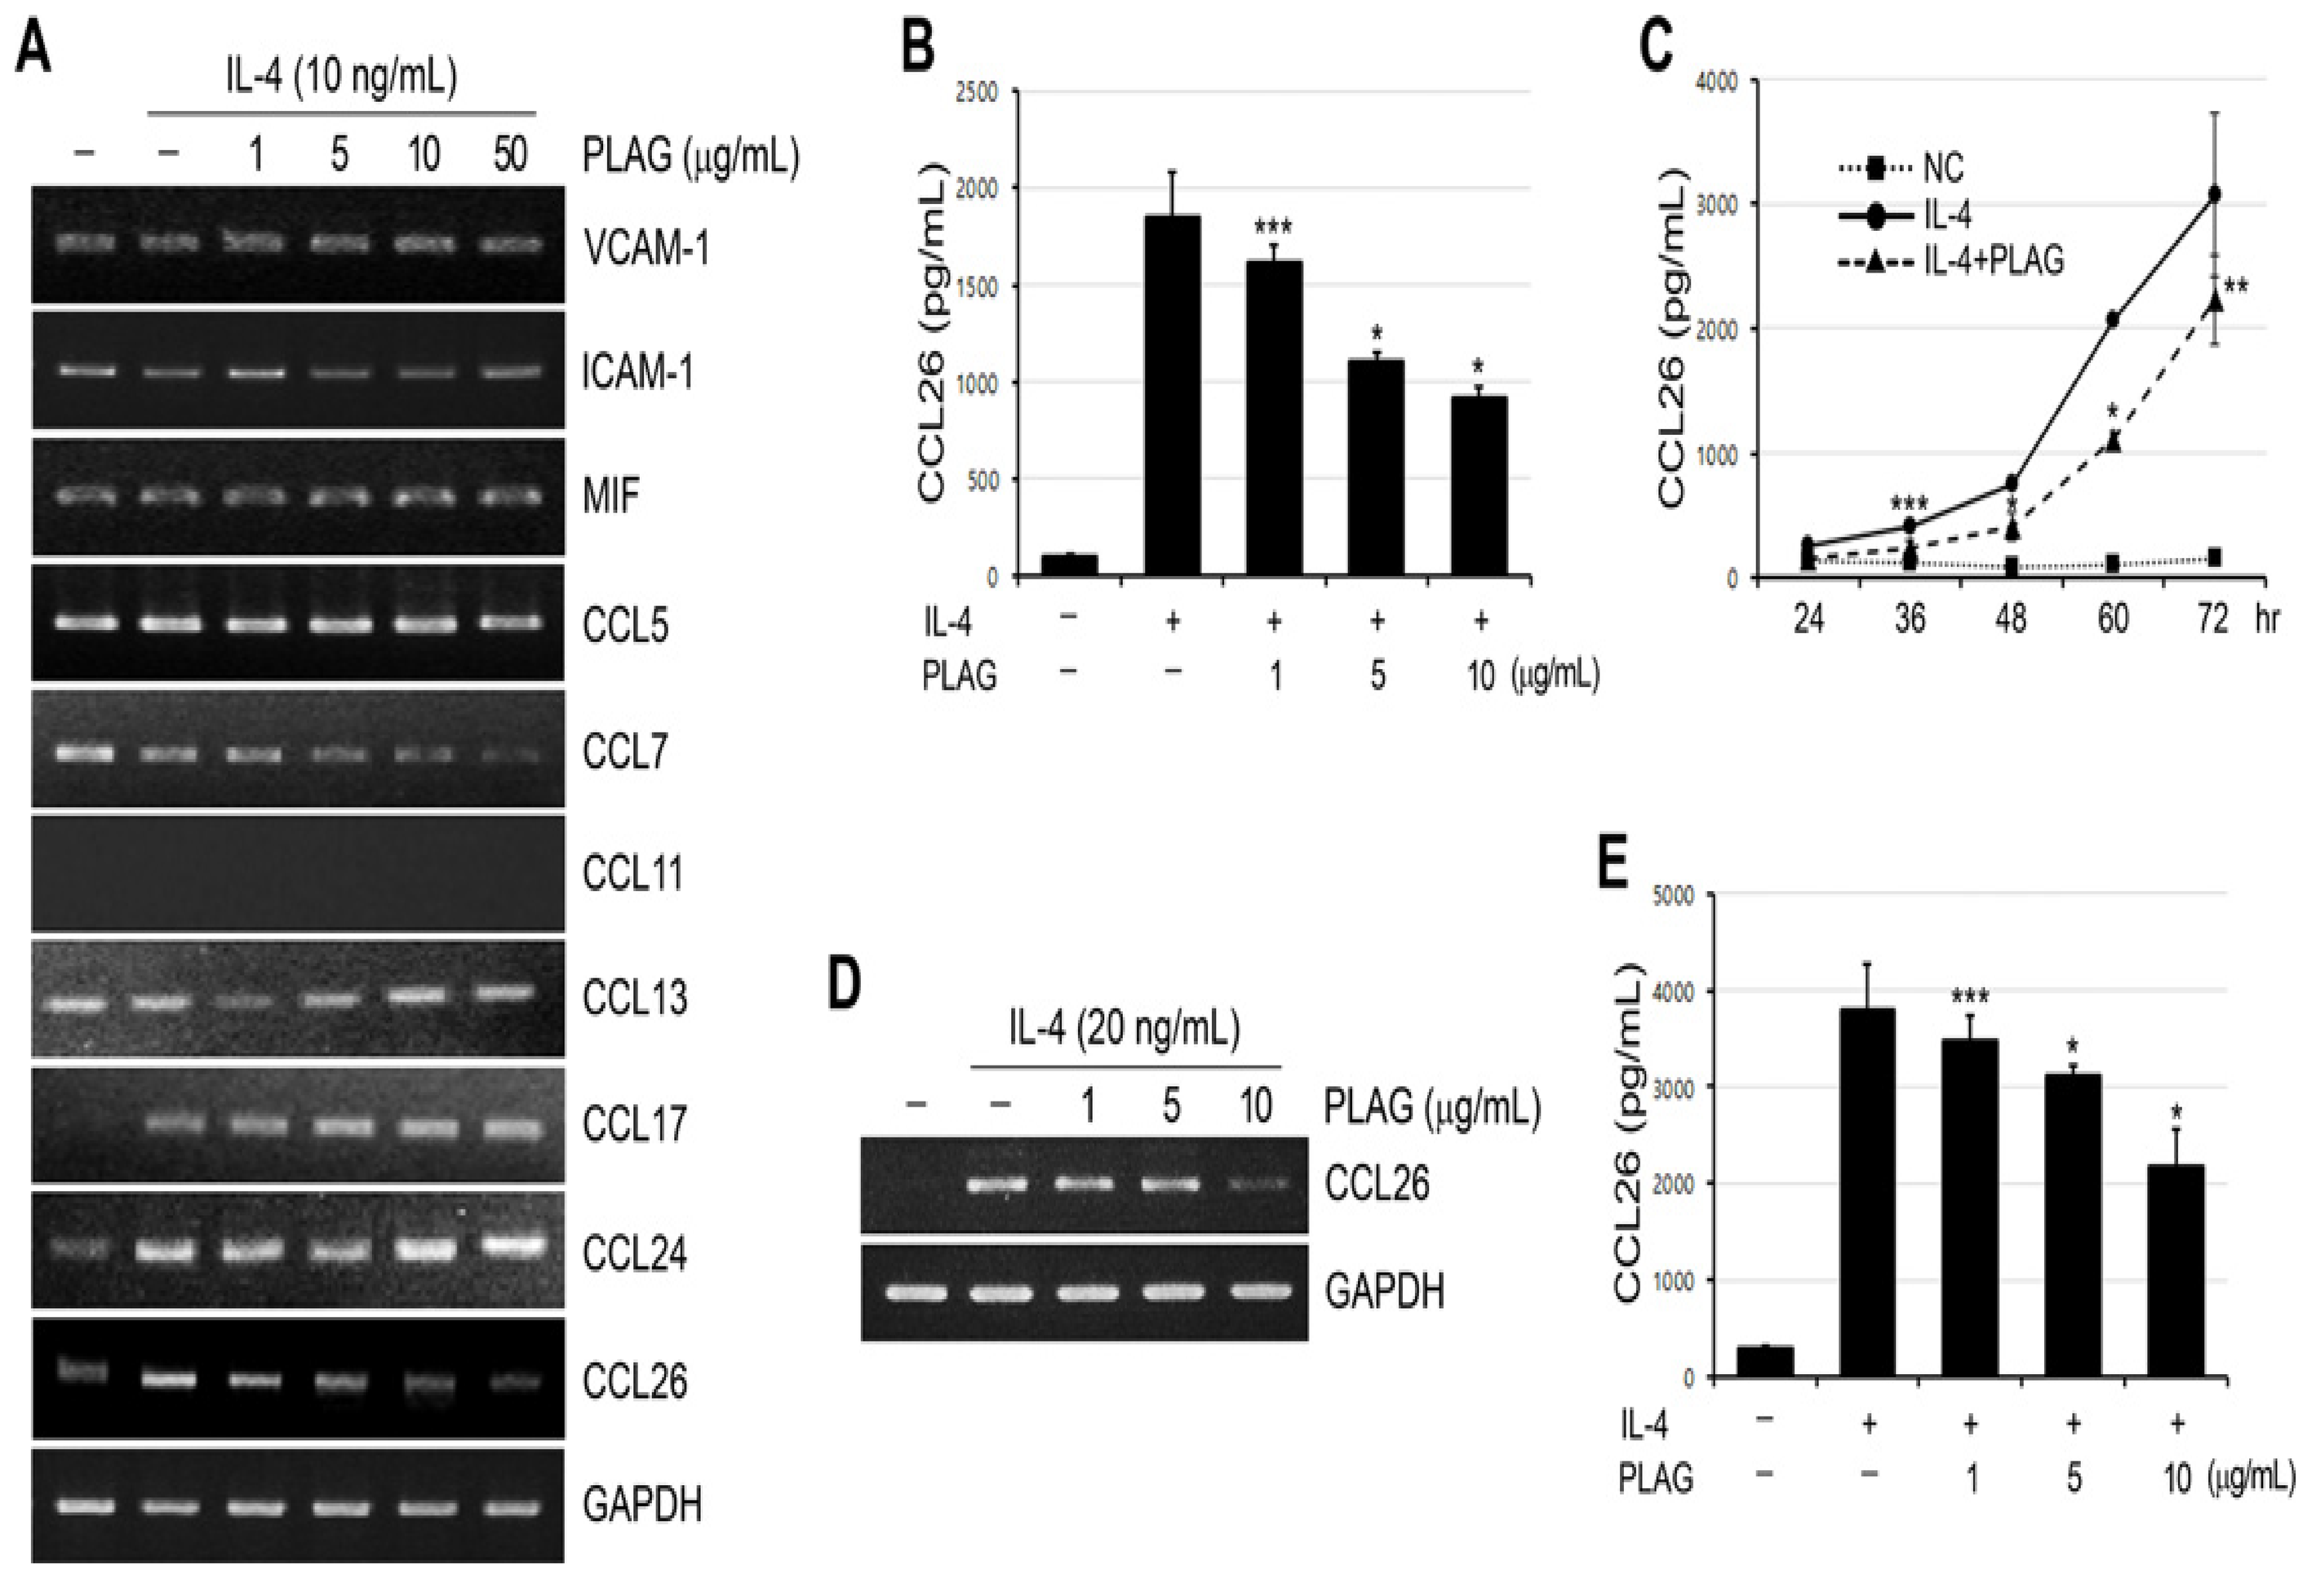

A
IL-4 (10 ng/mL)
PLAG (μg/mL)
VCAM-1
ICAM-1
MIF
CCL5
CCL7
CCL11
CCL13
CCL17
CCL24
CCL26
GAPDH
B
CCL26 (pg/mL)
C
NC
IL-4
IL-4+PLAG
hr
D
IL-4 (20 ng/mL)
E

Figure 1. (A) A549 cells were treated with various doses of PLAG for 1 h and stimulated with IL-4 (10 ng/mL) for 24 h. The mRNA levels of relevant genes were analyzed by RT-PCR. (B) The culture supernatant was harvested at 48 h, and the protein level of CCL26 was evaluated by ELISA. *p<0.001, ***p<0.05. (C) A549 cells were pretreated with PLAG (10 μg/mL) for 1 h and stimulated with IL-4 (10 ng/mL) for various lengths of time. NC; negative control. *p<0.001, **p<0.01, *** p<0.05. (D) HaCaT cells were treated with various doses of PLAG and stimulated with IL-4 (20 ng/mL) for 24 h. The mRNA levels of CCL 26 were analyzed by RT-PCR. (E) PLAG also decreased CCL26 secretion from HaCaT cells in a dose-dependent manner. *p<0.001, ***p<0.05.

2. PLAG inhibits transcriptional activity of STAT6.

It is well known that IL-4 induced CCL26 expression is mediated by JAK1/STAT6 signaling pathway. We confirmed that PLAG inhibits the transcriptional activity of STAT6 by regulating its phosphorylation and nuclear translocation.

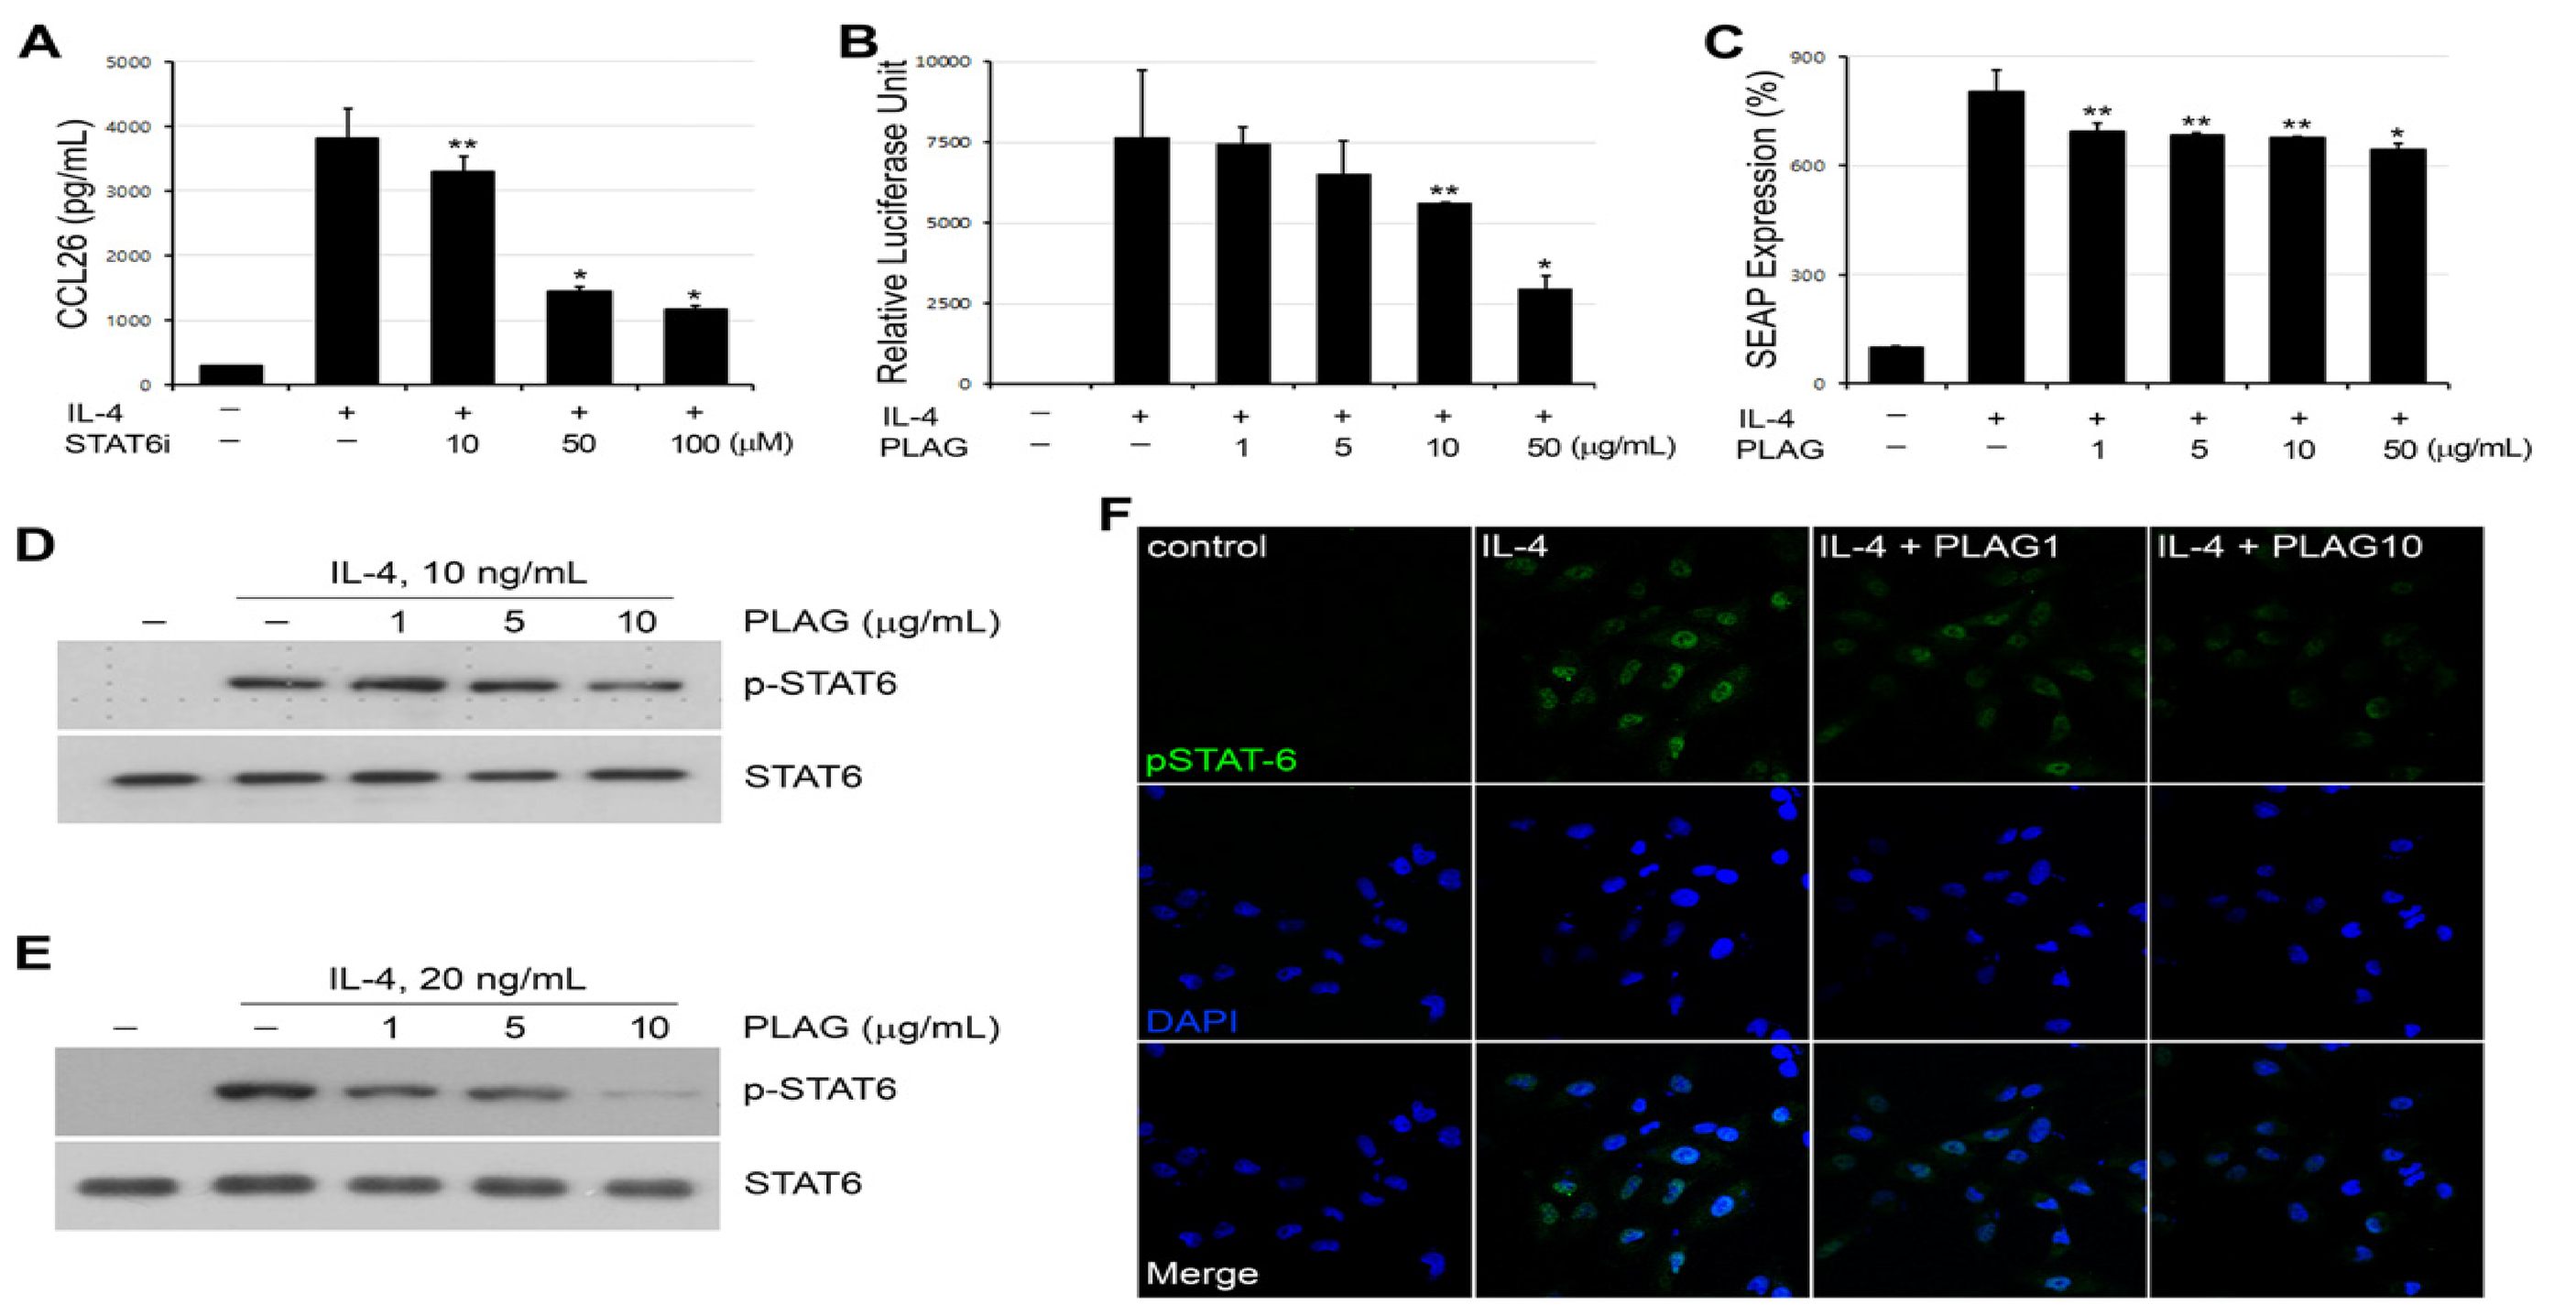

A
CCL26 (pg/mL)
IL-4
STAT6i
B
Relative Luciferase Unit
C
SEAP Expression (%)
D
IL-4, 10 ng/mL
PLAG (μg/mL)
p-STAT6
STAT6
E
IL-4, 20 ng/mL
F
control
IL-4
IL-4 + PLAG1
IL-4 + PLAG10
pSTAT-6
DAPI
Merge

Figure 2. (A) Various concentration of a STAT6 inhibitor (STAT6i, AS1517499) were treated onto A549 cells which were activated by IL-4 (10 ng/mL), and showed inhibitory effect on the secretion of CCL26 from the epithelial cells in a concentration-dependent manner. *p<0.001, **p<0.01. (B) Reporter construct containing luciferase gene regulated by STAT6 activity was transfected into A549 cells and the effect of PLAG on the expression of the luciferase gene was analyzed by Dual-Glo luciferase system. The inhibitory effect of PLAG on STAT6 activity resulted in the decrease of luciferase expression. *p<0.001, **p<0.01. (C) The effect of PLAG on the inhibition of IL-4-induced SEAP activity was evaluated in HEK Blue IL-4/IL-13 cells. *p<0.001, **p<0.01. (D-E) Cellular extracts of IL-4 and/or PLAG treated epithelial cells were analyzed by Western blotting. PLAG decreased the phosphorylation of STAT6 with dose dependency in both A549 (D) and HaCaT (E). (F) Confocal microscopy showed that IL-4 induced nuclear localization of phosphorylated STAT6, which was inhibited by the co-treatment of PLAG.

3. PLAG alleviates inflammatory phenotypes in allergic asthma model.

A large number of inflammatory cells including eosinophils were infiltrated into the lung tissue of OVA-challenged mice group compared with the control group. PLAG administration significantly reduced the infiltration of immune cells similarly with DEX treatment. CCL26 concentration in BALF was also effectively reduced by DEX or PLAG administration.

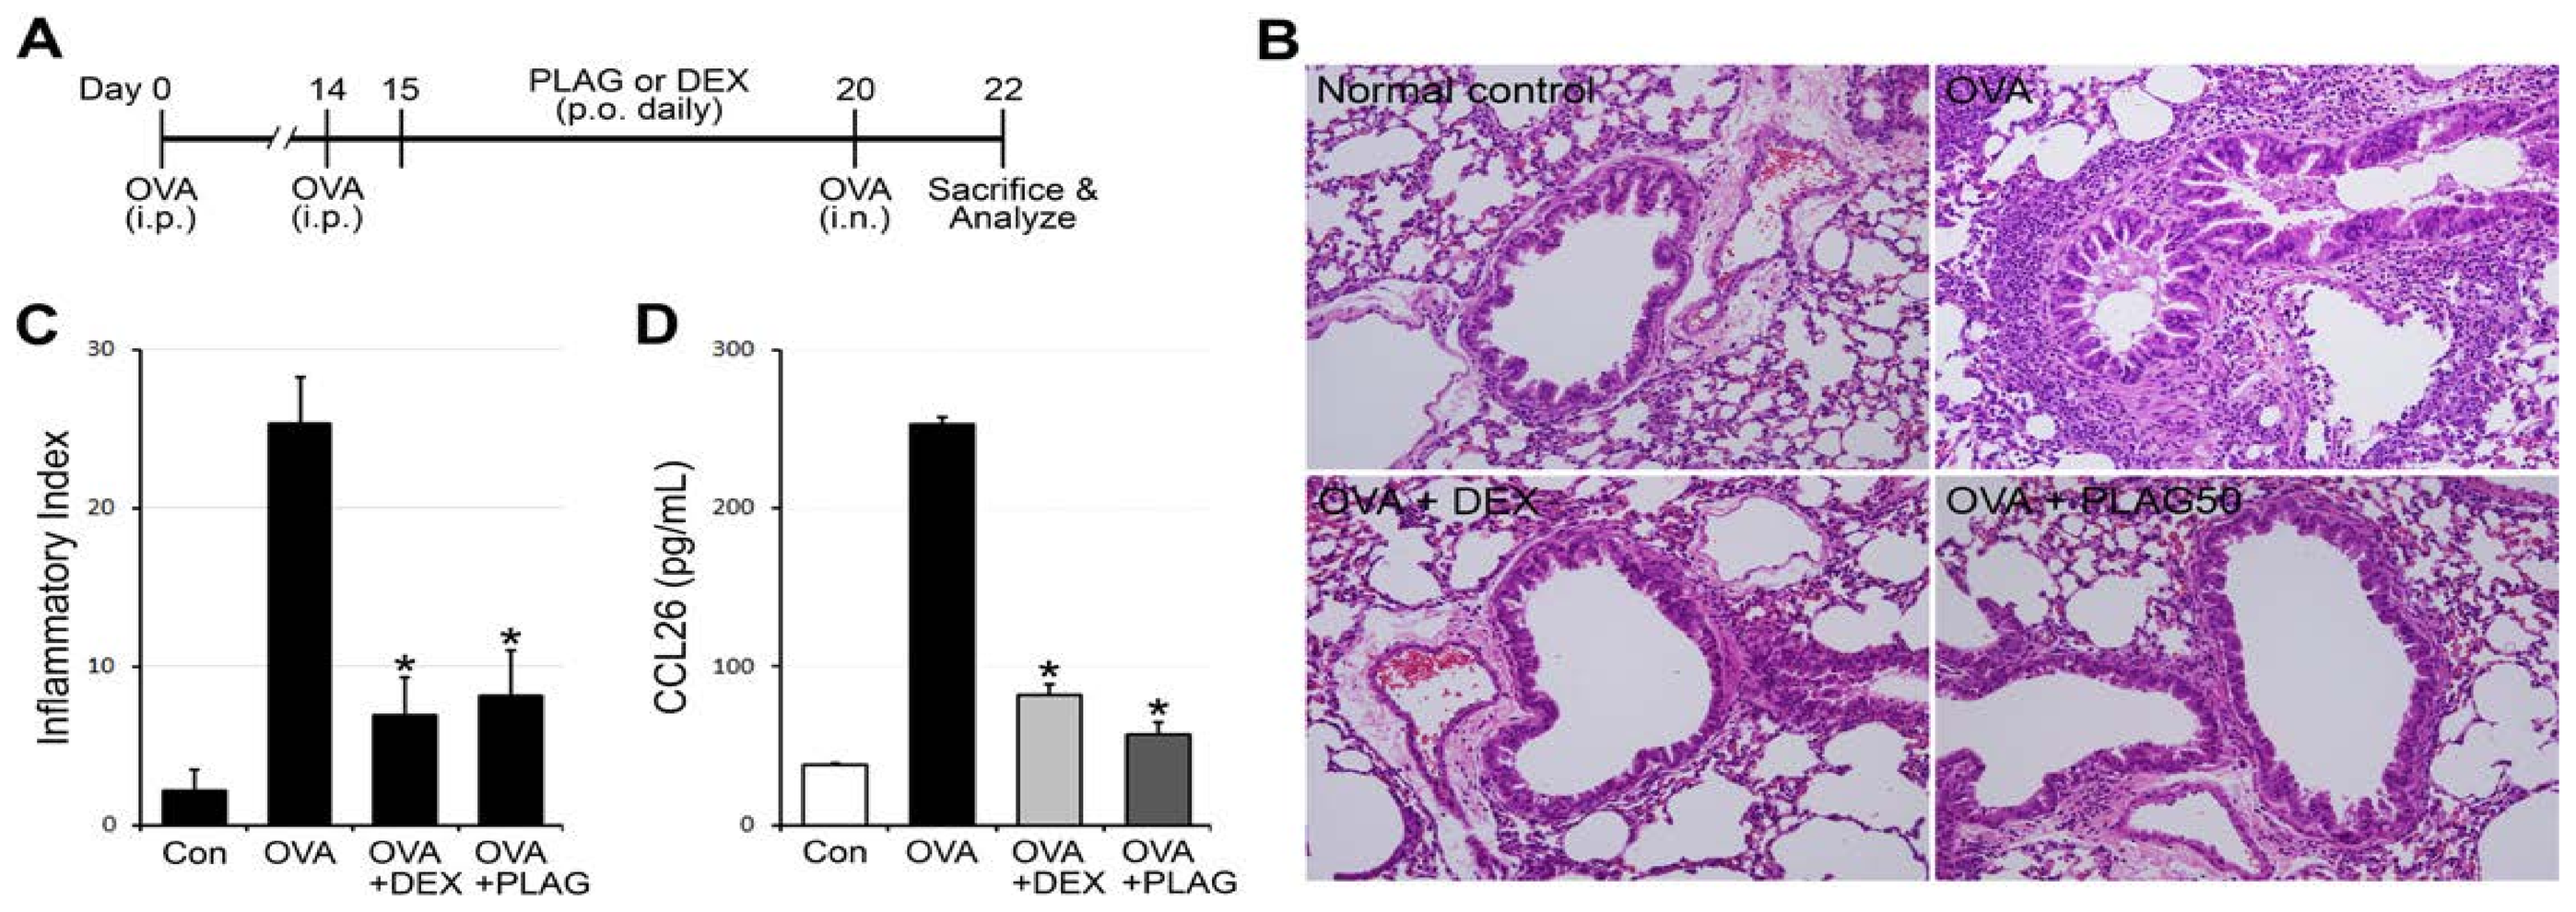

A
Day 0
14
15
PLAG or DEX (p.o. daily)
20
22
OVA (i.p.)
OVA (i.p.)
OVA (i.n.)
Sacrifice & Analyze
B
Normal control
OVA
OVA + DEX
OVA + PLAG50
C
Inflammatory Index
D
CCL26 (pg/mL)
Con
OVA
OVA +DEX
OVA +PLAG

Figure 3. (A) Mice were sensitized and challenged by OVA, and the efficacy of PLAG and DEX was tested in the generated asthma model as depicted. (B-C) The lung tissues from the OVA-sensitized and–challenged mice were stained with H&E, and the representative images were displayed (X200 magnification, B). Infiltrated immune cells were counted, calculated and displayed as inflammation index. Eosinophils from each of the three mice per treatment group were analyzed morphometrically and mean eosinophil counts per 50 × 50 μm area ± SD were displayed. Four such areas were measured from each tissue section. PLAG was the most effective with a dose of 50 mg/Kg as shown in (B) and calculated in (C). *p<0.001. (D) The expression level of CCL26 in BALFs of OVA-challenged mice was analyzed by ELISA. Each sample was analyzed in triplicate and displayed as mean ± SD. *p<0.001.

4. PLAG decreases eosinophil infiltration into the airways of asthmatic mice.

CBC analysis of the BALFs showed that most blood cell populations were massively increased in OVA-challenged mice, which were decreased by PLAG and DEX treatment. Especially, the number of infiltrated eosinophils into the airway of OVA-challenged mice was decreased in PLAG administered mice, as seen in CBC and FACs analysis.

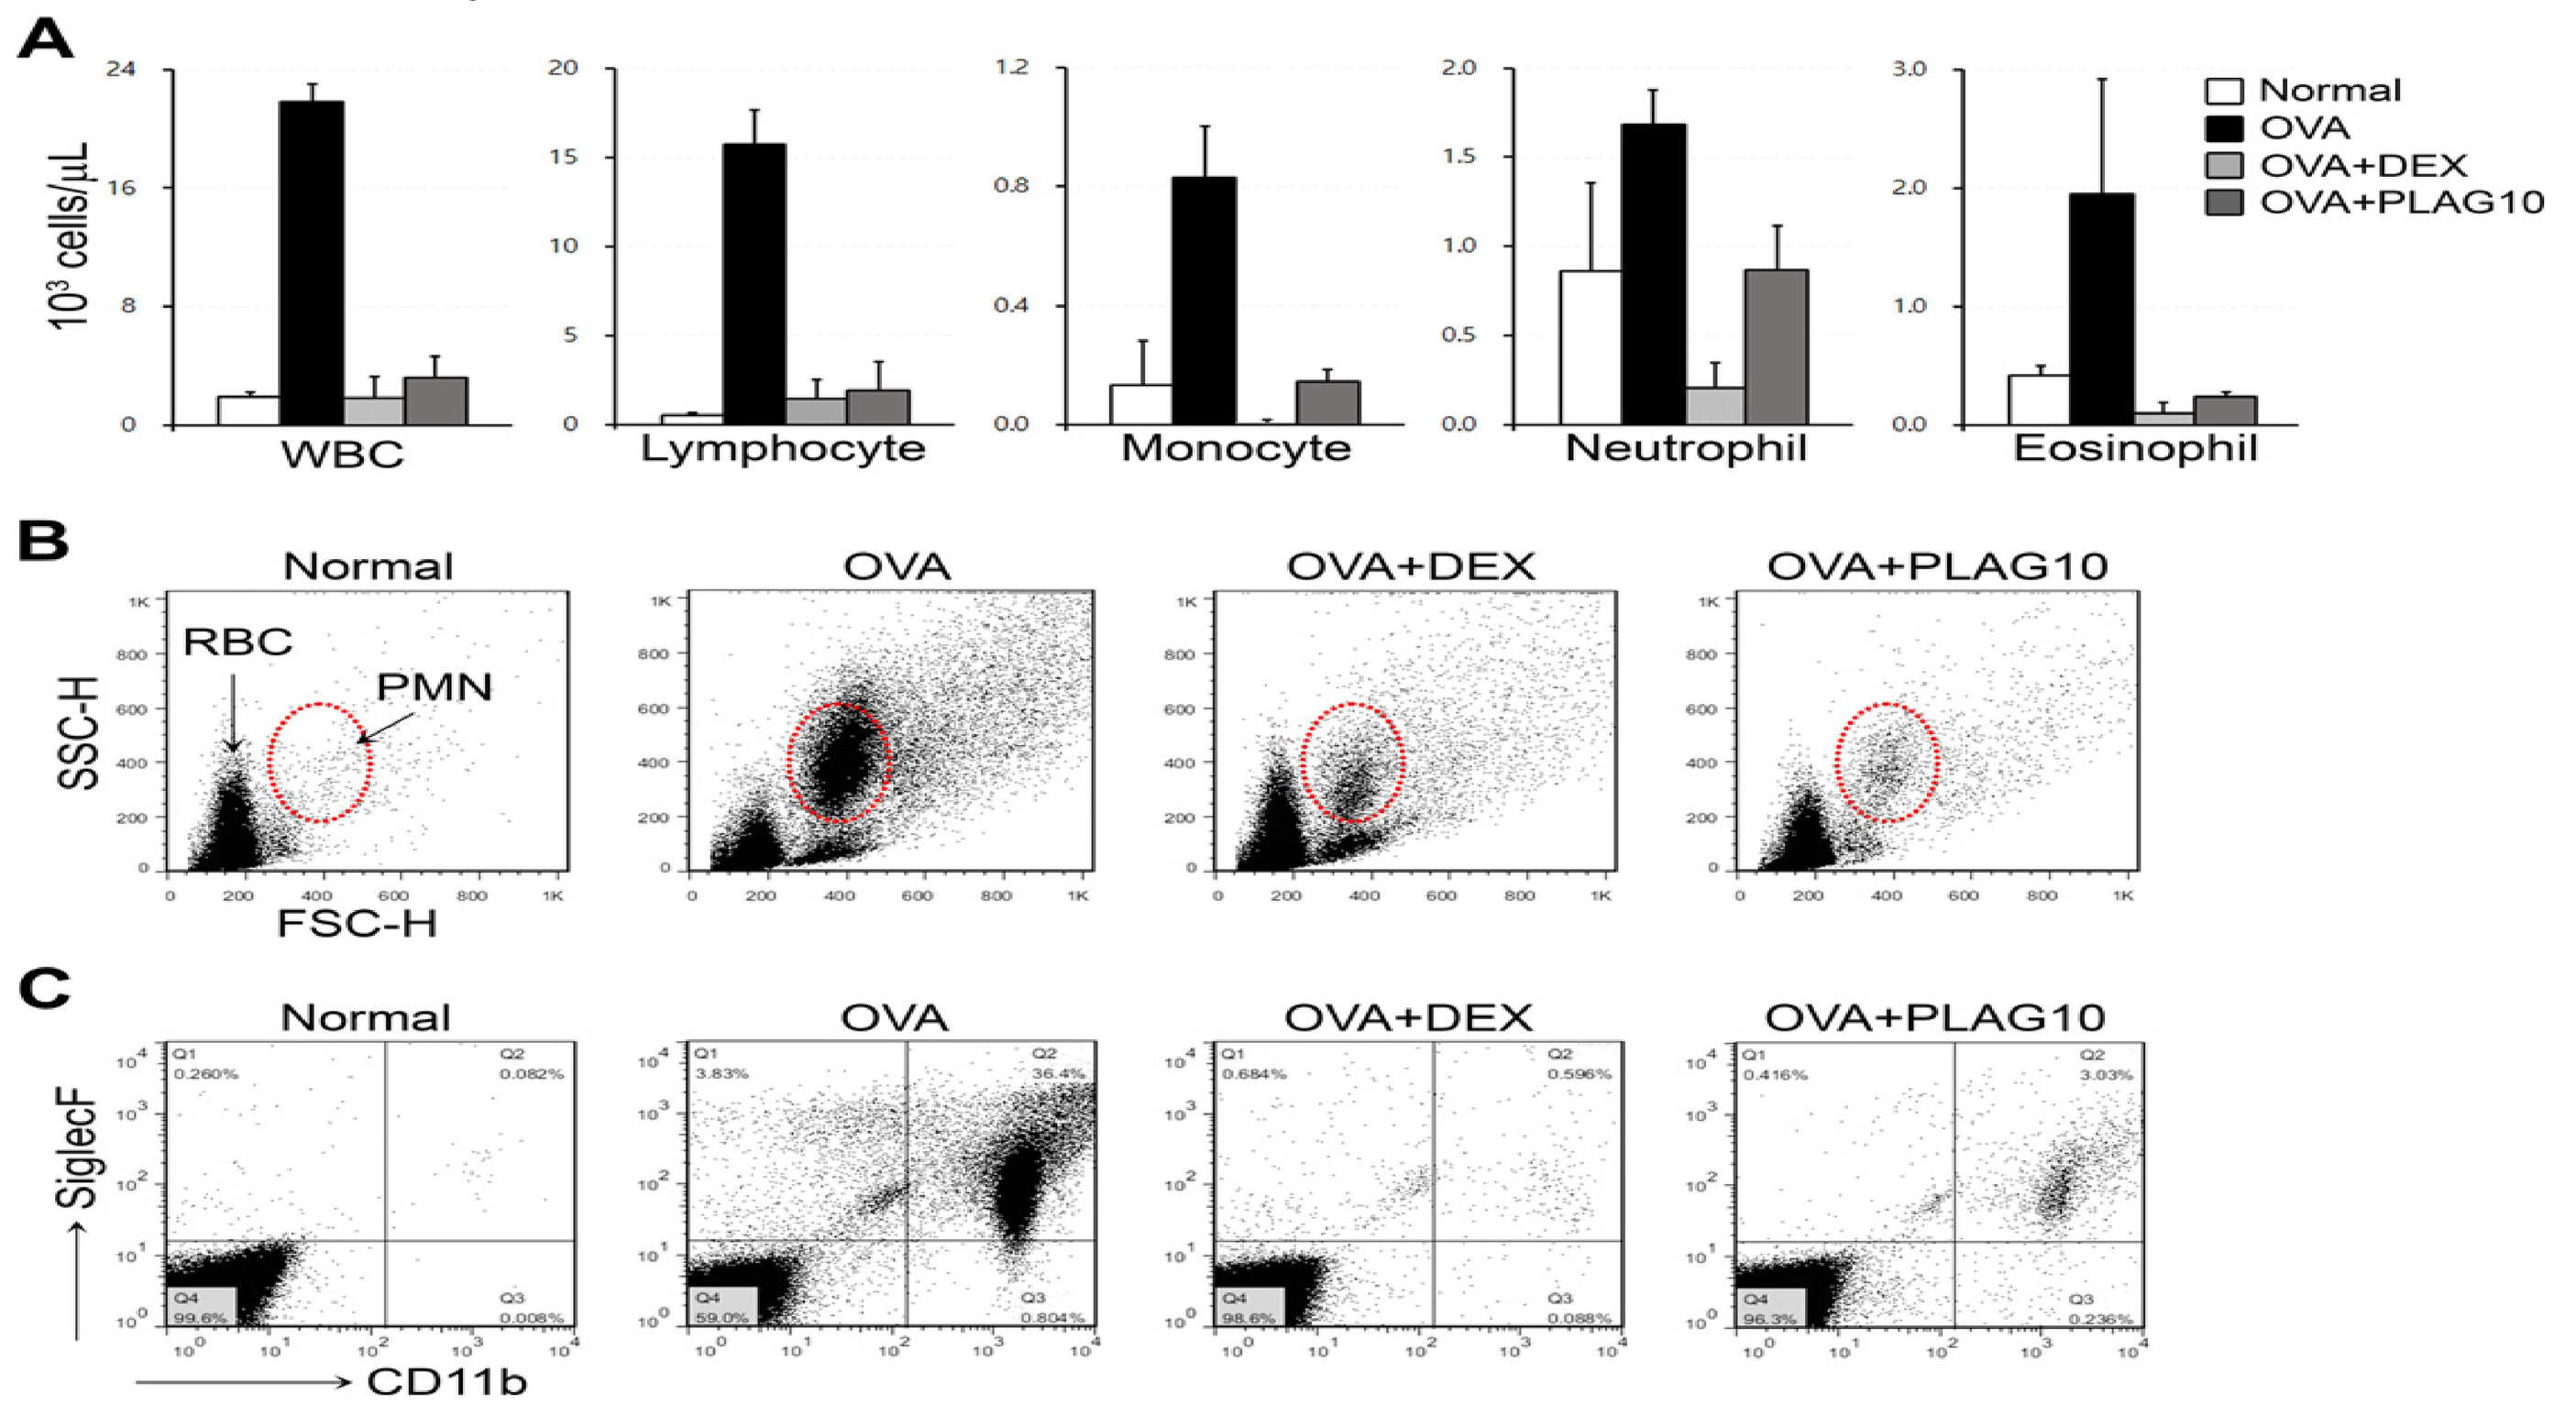

A
10³ cells/μL
WBC
Lymphocyte
Monocyte
Neutrophil
Eosinophil
Normal
OVA
OVA+DEX
OVA+PLAG10
B
RBC
PMN
SSC-H
FSC-H
C
SiglecF
CD11b

Figure 4. (A) Cells were harvested from the BALFs of OVA-challenged mice and analyzed by CBC. The number of infiltrated immune cells were increased in OVA-challenged mice. PLAG or DEX treatment inhibited immune cell infiltration into airways to the level of normal control. (B) Cells were collected from the BALF, and analyzed by flow cytometry. (C) SiglecF+/CD11b+ eosinophils were increased in the OVA-challenged mice, which were decreased in PLAG- or DEX-treated mice.

5. PLAG inhibits eosinophil migration by suppressing CCL26 expression in vitro.

We performed chemotaxis assay using murine eosinophils differentiated from bone marrow cells and a human eosinophil cell line, EoL-1. CCL26-containing medium from IL-4 stimulated A549 cells caused chemotactic invasion of these cells, which was inhibited by pre-treatment of PLAG in a concentration dependent manner.

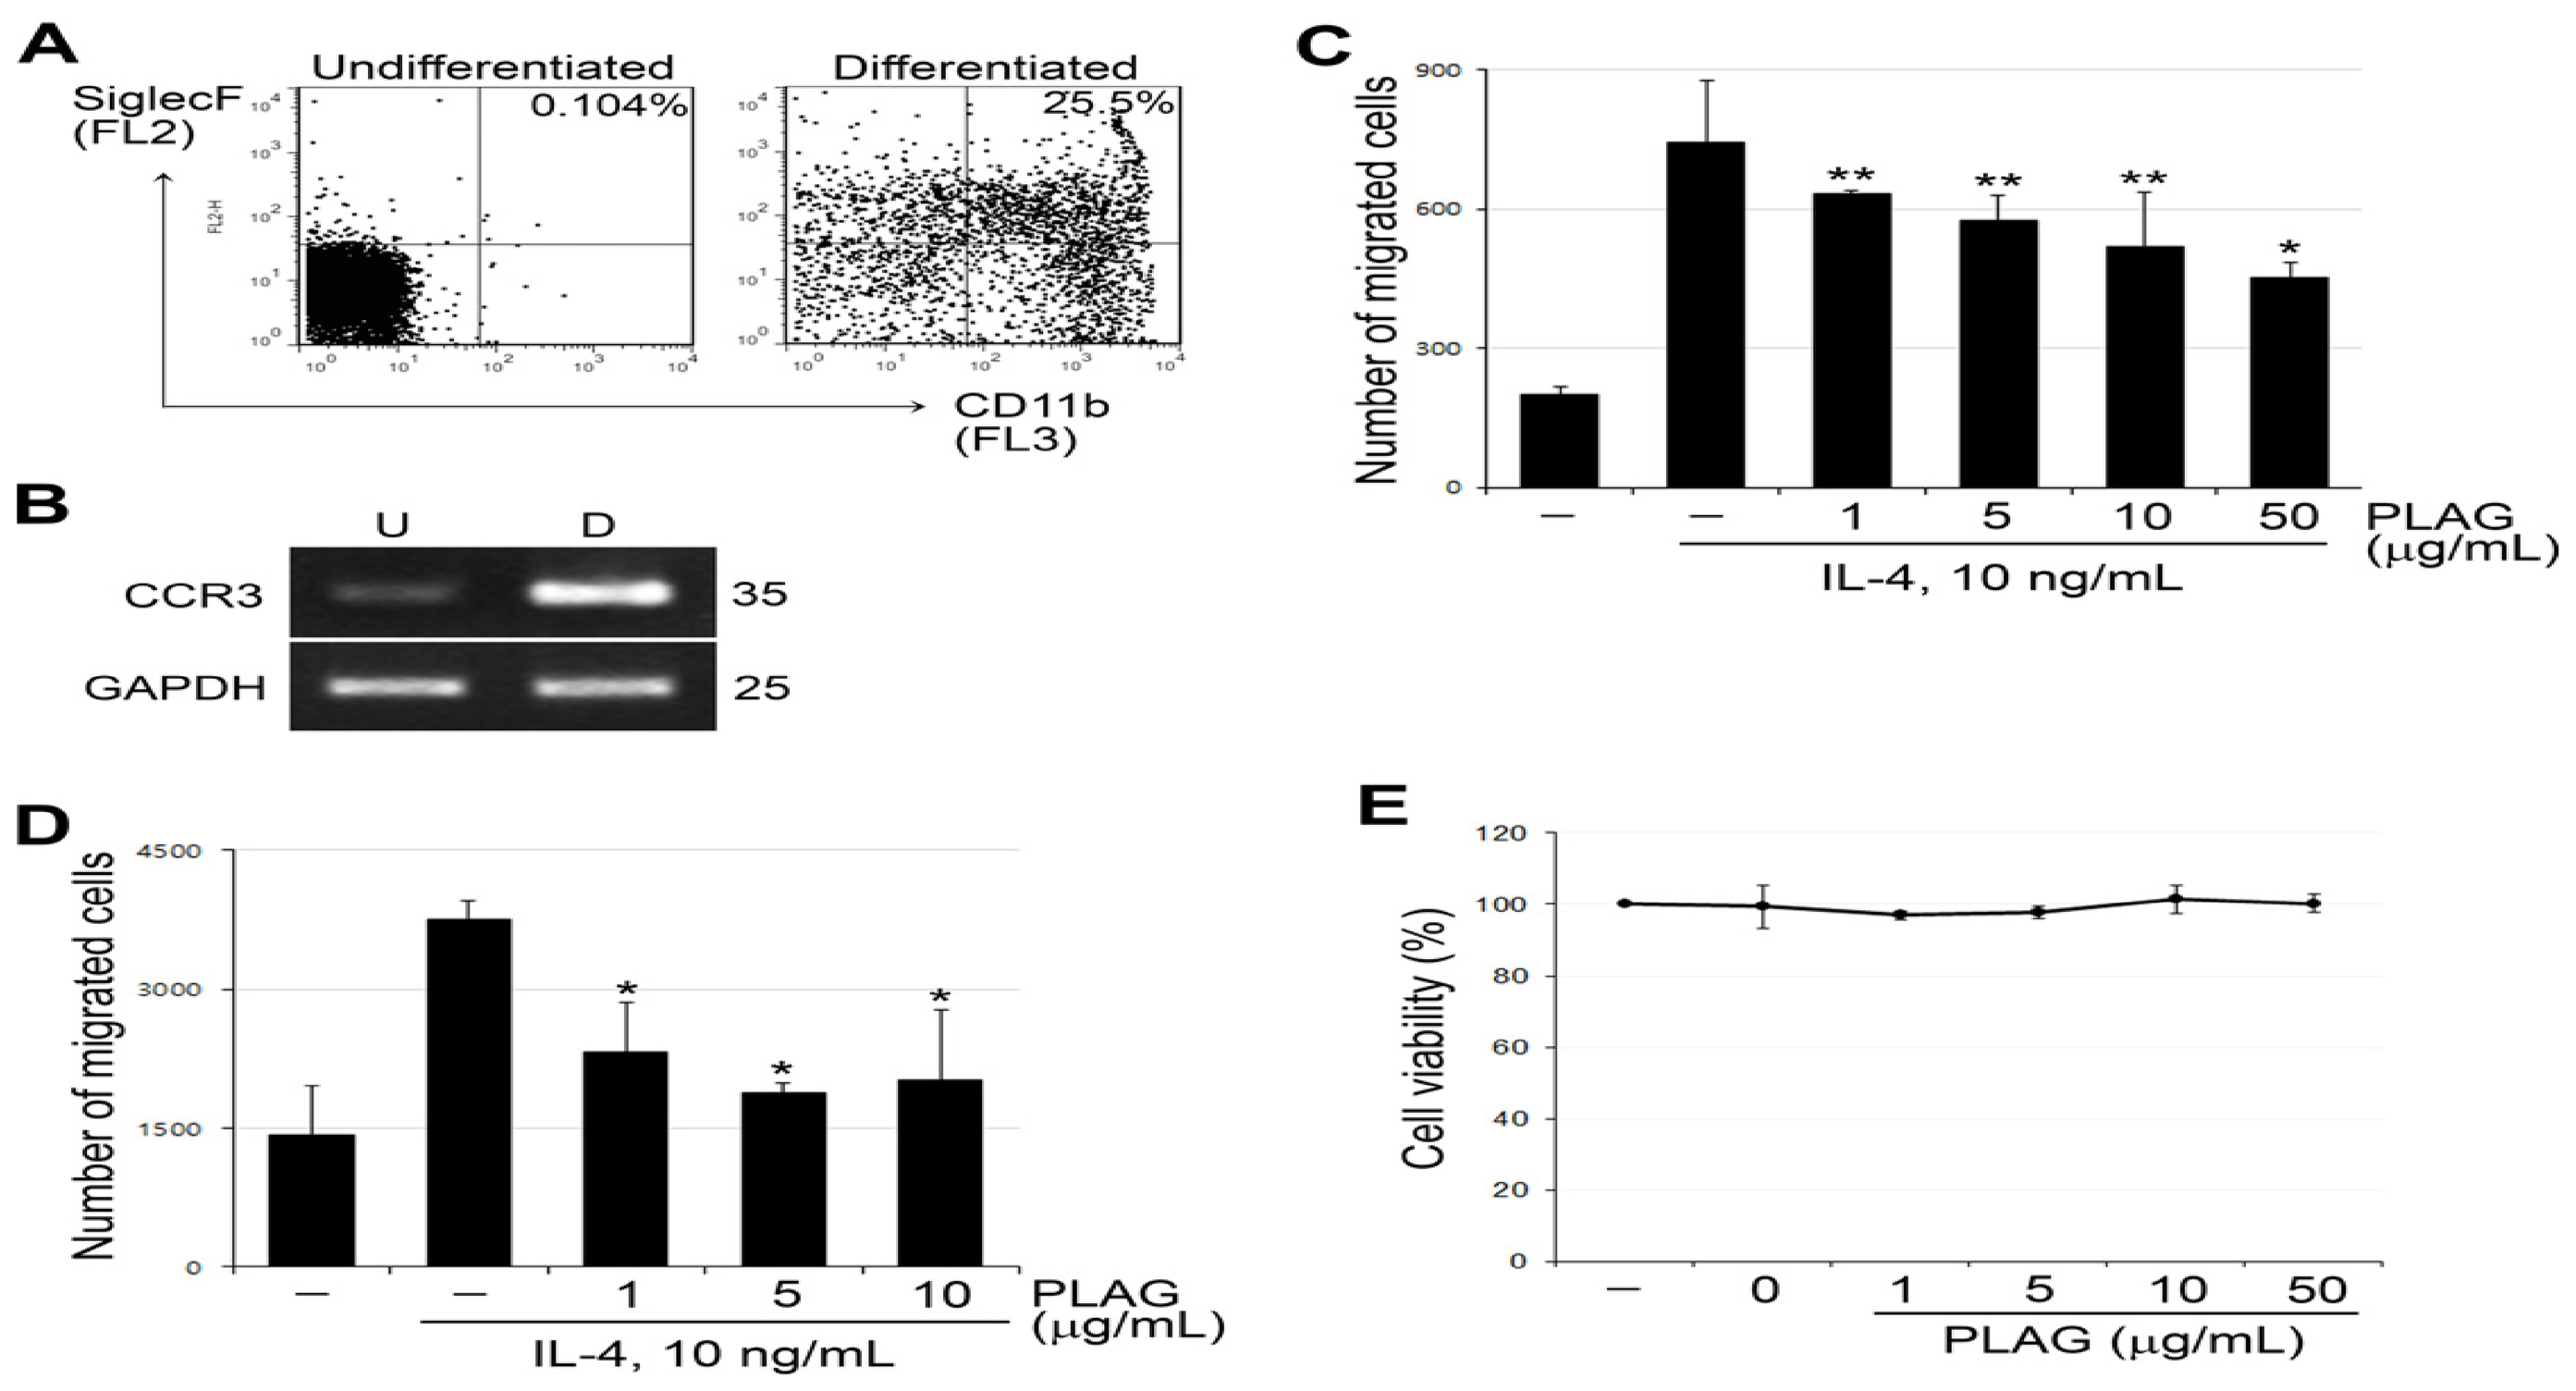

A
SiglecF (FL2)
Undifferentiated
0.104%
Differentiated
25.5%
CD11b (FL3)
B
U
D
CCR3
GAPDH
35
25
C
Number of migrated cells
PLAG (μg/mL)
IL-4, 10 ng/mL
D
E
Cell viability (%)
PLAG (μg/mL)

Figure 5. (A) Murine eosinophils were differentiated from BM cells by treating SCF, FLT3 ligand and IL-5 for 2w. The differentiated cells were analyzed by FACS staining with SiglecF and CD11b. SiglecF+/CD11b+ cells were increased in the differentiated cells to ~25%. (B) CCR3 expression was increased in the differentiated murine eosinophils. (C-D) The supernatant of IL-4 and/or PLAG-treated A549 cells was prepared and used for the migration assay. The transmigration of eosinophil was decreased by PLAG treatment in both murine BM-derived eosinophils (C) and human eosinophil cell line, EoL-1 (D). *p<0.001, **p<0.01. (E) PLAG did not exhibit any cytotoxic effects on A549 cells at various concentrations.

Conclusion

- Infiltrated eosinophils into the airway are an important phenotype of asthma. Treatment of PLAG significantly reduced the number of immune cell infiltration including eosinophils in the airway in the OVA-induced allergic asthma mice model.
- Epithelial cells produced CCL26 in response to IL-4 stimulation, and PLAG effectively attenuated mRNA and protein expression of CCL26 induced by IL-4 through regulating the transcriptional activity of STAT6.
- PLAG inhibited eosinophil transmigration in vitro system by down-regulating CCL26 secretion in the supernatant of IL-4 stimulated A549 cells.

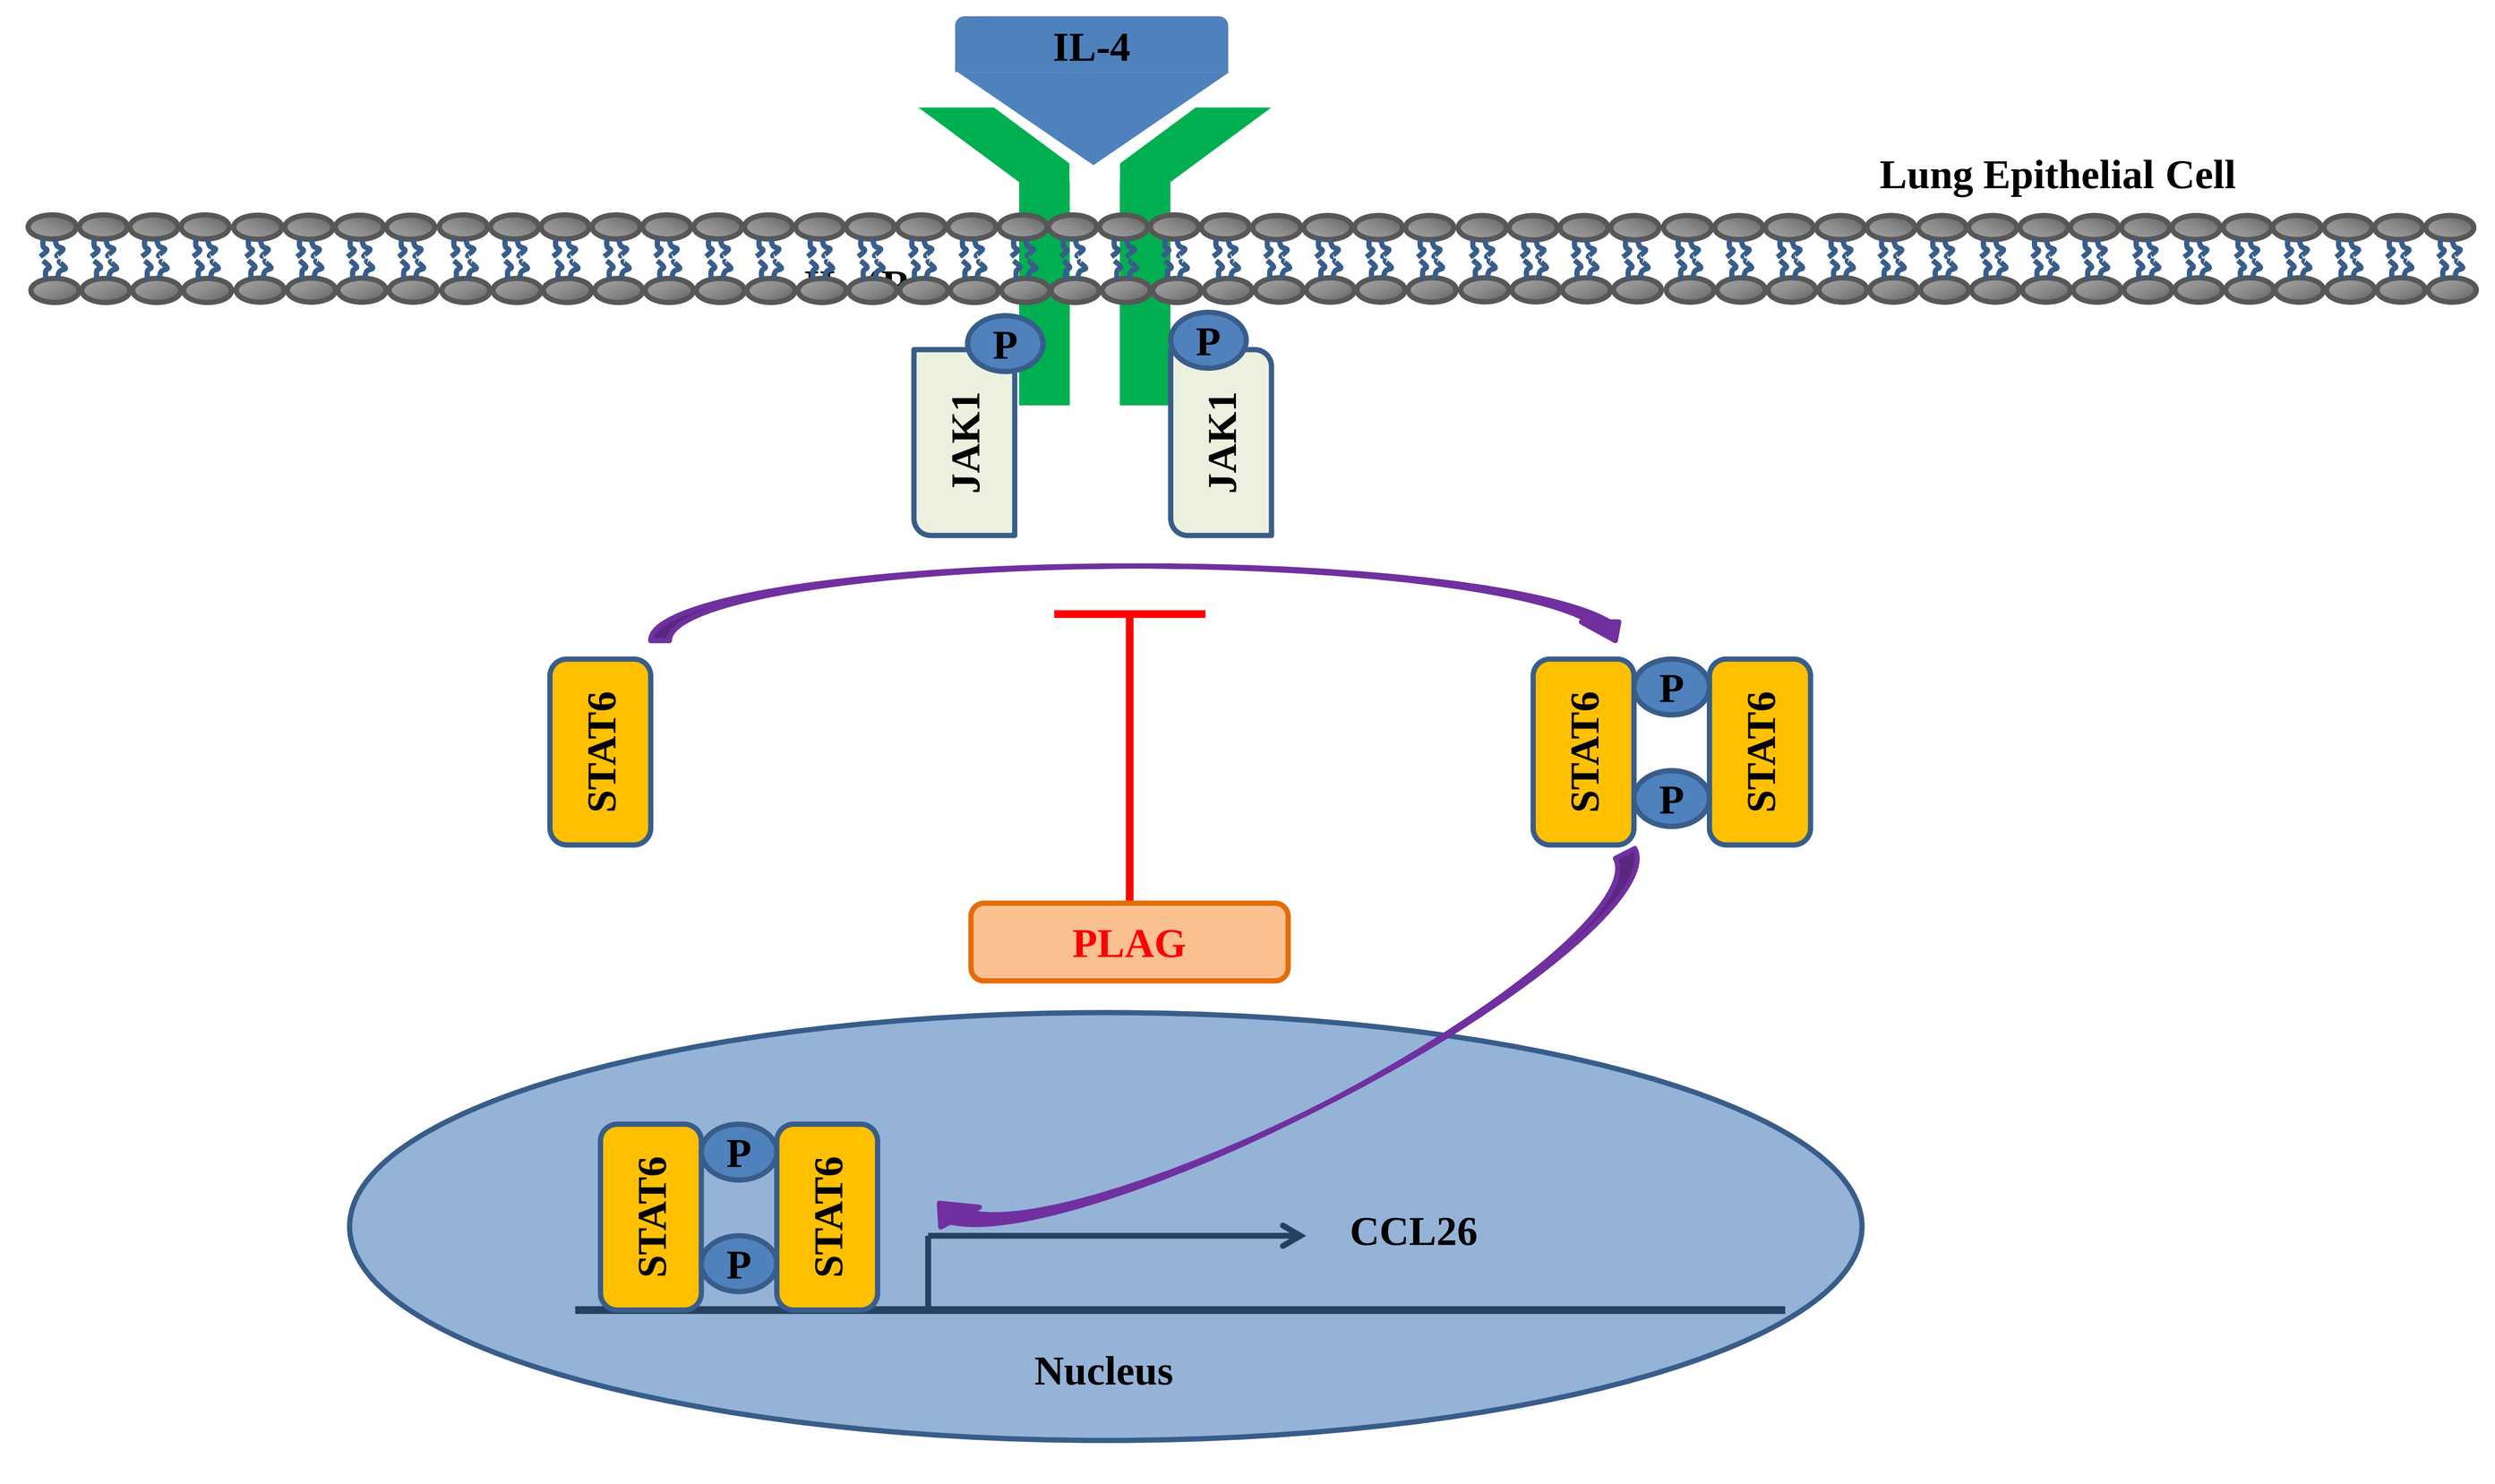

IL-4
Lung Epithelial Cell
P
JAK1
STAT6
PLAG
CCL26
Nucleus

Diagram of inhibitory effects of EC-18 on IL-4 induced CCL26 production

References

[1] Lambrecht BN., *et al*. (2015). **Nat Immunol.** 16(1):45–56.

[2] Kay AB., et al. (2004). **Trends Immunol.** 25(9):477–82.

[3] Banwell ME., *et al*. (2002). **Cytokine.** 17(6): 317-23.

[4] Hoeck J., *et al*. (2001). **J Immunol.** 167(6):3216-22.

[5] Yoon, S. Y., et al. (2015). **Immune Netw.** 15(2): 100-109.