Gemcitabine-generated ROS promotes neutrophil chemotactic activity through CXCL8 production via PKCδ/ERK pathway in a murine 4T1 breast cancer model

Jinseon Jeong[1,2,3], Yong-Jae Kim[1], Kwang Hoon Yang[1,4], Sun Young Yoon[3], Ki-Young Sohn[3], Heung-Jae Kim[3] and Jae Wha Kim[1,2]

[1]Cell Factory Research Center, Korea Research Institute of Bioscience and Biotechnology, Republic of Korea. [2] Department of Functional Genomics, KRIBB School of Bioscience, University of Science & Technology, Daejeon 34113, Republic of Korea. [3]Enzychem Lifesciences Corporation, F741 KAIST-ICC, Daejeon 305-732, Republic of Korea. [4]Chonnam National University, Republic of Korea.

P672

Abstract

Chemotherapy-induced neutropenia (CIN) is a common side effect that necessitate dose reductions during treatment of cancer patients. Preventing CIN is critical in chemotherapy because a rapid decline of neutrophil counts increase susceptibility to infection of cancer patients. In spite of its importance, the mechanism of CIN still remains unclear. This study is aimed to investigate the mechanism of CIN in an aspect of neutrophil activation by chemotaxis in gemcitabine-treated breast tumor model. We hypothesized that gemcitabine-generated ROS promotes CXCL8 or MIP-2 production by upregulating ERK pathway in PKCδ-activated intra-tumoral macrophages and thus neutrophil recruitment to the chemokine gradient. In 4T1 breast tumor mouse model, we found that the basal level of PKCδ activity was higher than in normal mice. The number of blood and peritoneal leukocytes was analyzed at 24h after intraperitoneal administration of gemcitabine. The neutrophil counts decreased in the blood but increased in the peritoneal cavity. In addition, intratumoral neutrophils were elevated in the gemcitabine-treated group. The mRNA levels of neutrophil attracting chemokines/receptors were increased in the peritoneal and tumoral cells. ROS inhibitors N-Acetyl-L-cysteine and Apocynin blocked gemcitabine-induced neutrophil migration, mRNA expression of the chemokines and receptors and PKCδ/ERK phosphorylation. CXCR1/2 inhibitor reparixin decreased neutrophil recruitment to the peritoneal cavity and the tumor tissue. Together, our results suggest that gemcitabine treatment promotes neutrophil chemotactic activity by increasing ROS/PKCδ/ERK/CXCL8 pathway. These findings will provide new insights for developing a therapeutic for CIN.

Introduction

- Chemotherapy-Induced Neutropenia (CIN) is a common side effect observed in chemotherapy treated cancer patients [1].
- Neutrophils are circulating granulocytes responsible for a first line of defenses against pathogen infections [2]. Recent studies suggest that tumor-associated neutrophils are closely associated with caner initiation and progression by producing cytokines and promoting angiogenesis [3].
- Gemcitabine is a DNA analog type of chemotherapeutic agent that induces cell deaths by interfering with DNA synthesis and repair [4].
- Chemotherapeutic agents generate reactive oxygen species (ROS) production by activating NADPH oxidase which leads to the activation of PKCδ and ERK1/2 MAPK [5].
- CXCL8 is a primary chemokine involved in the recruitment of neutrophils into damaged tissues and mainly produced by monocytes/macrophages [2].

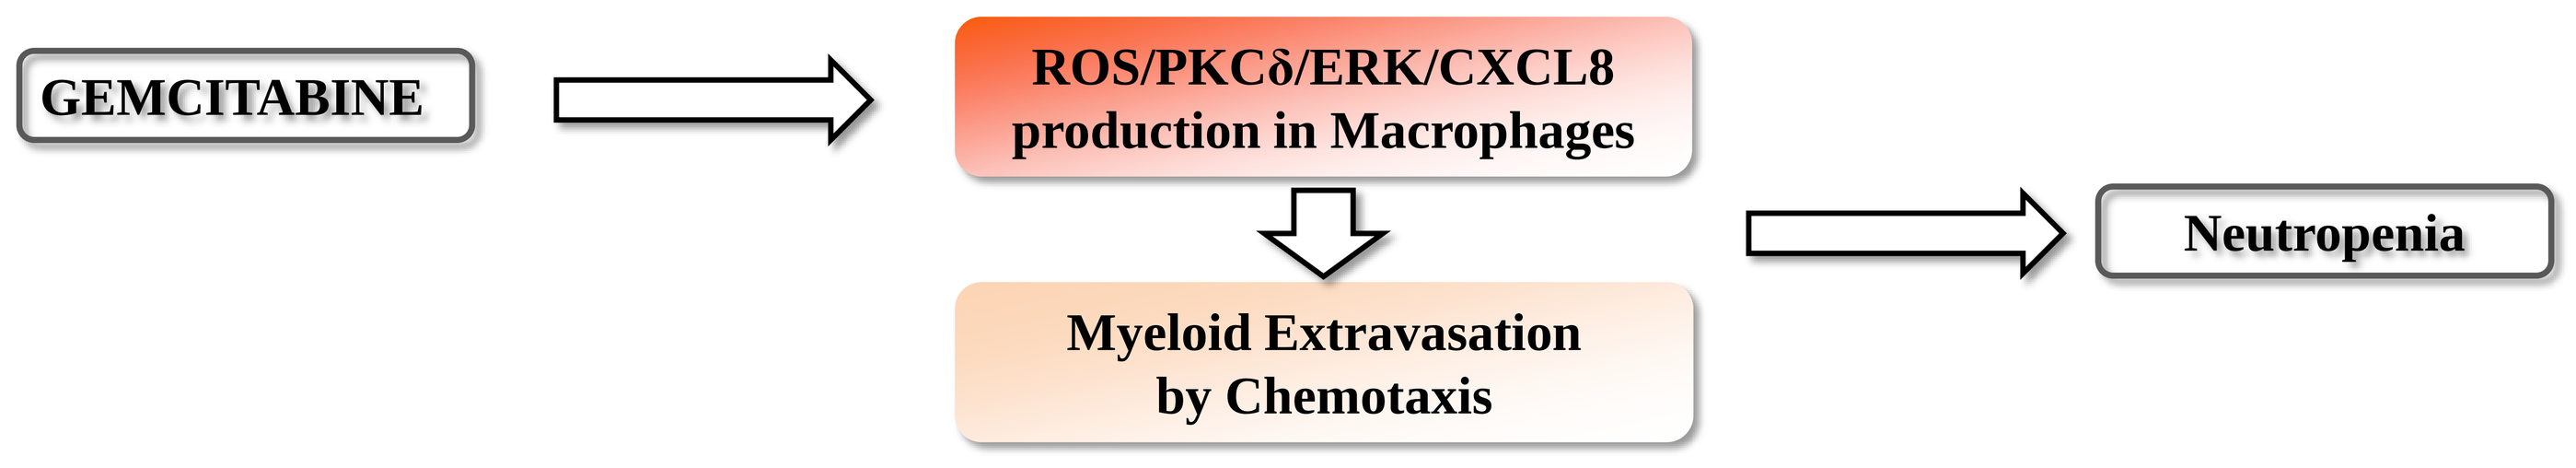
GEMCITABINE
ROS/PKCδ/ERK/CXCL8 production in Macrophages
Myeloid Extravasation by Chemotaxis
Neutropenia

Diagram of CIN (chemotherapy-induced neutropenia)

Results

1. Gemcitabine induces neutropenia by promoting neutrophil transmigration into the peritoneal cavity.

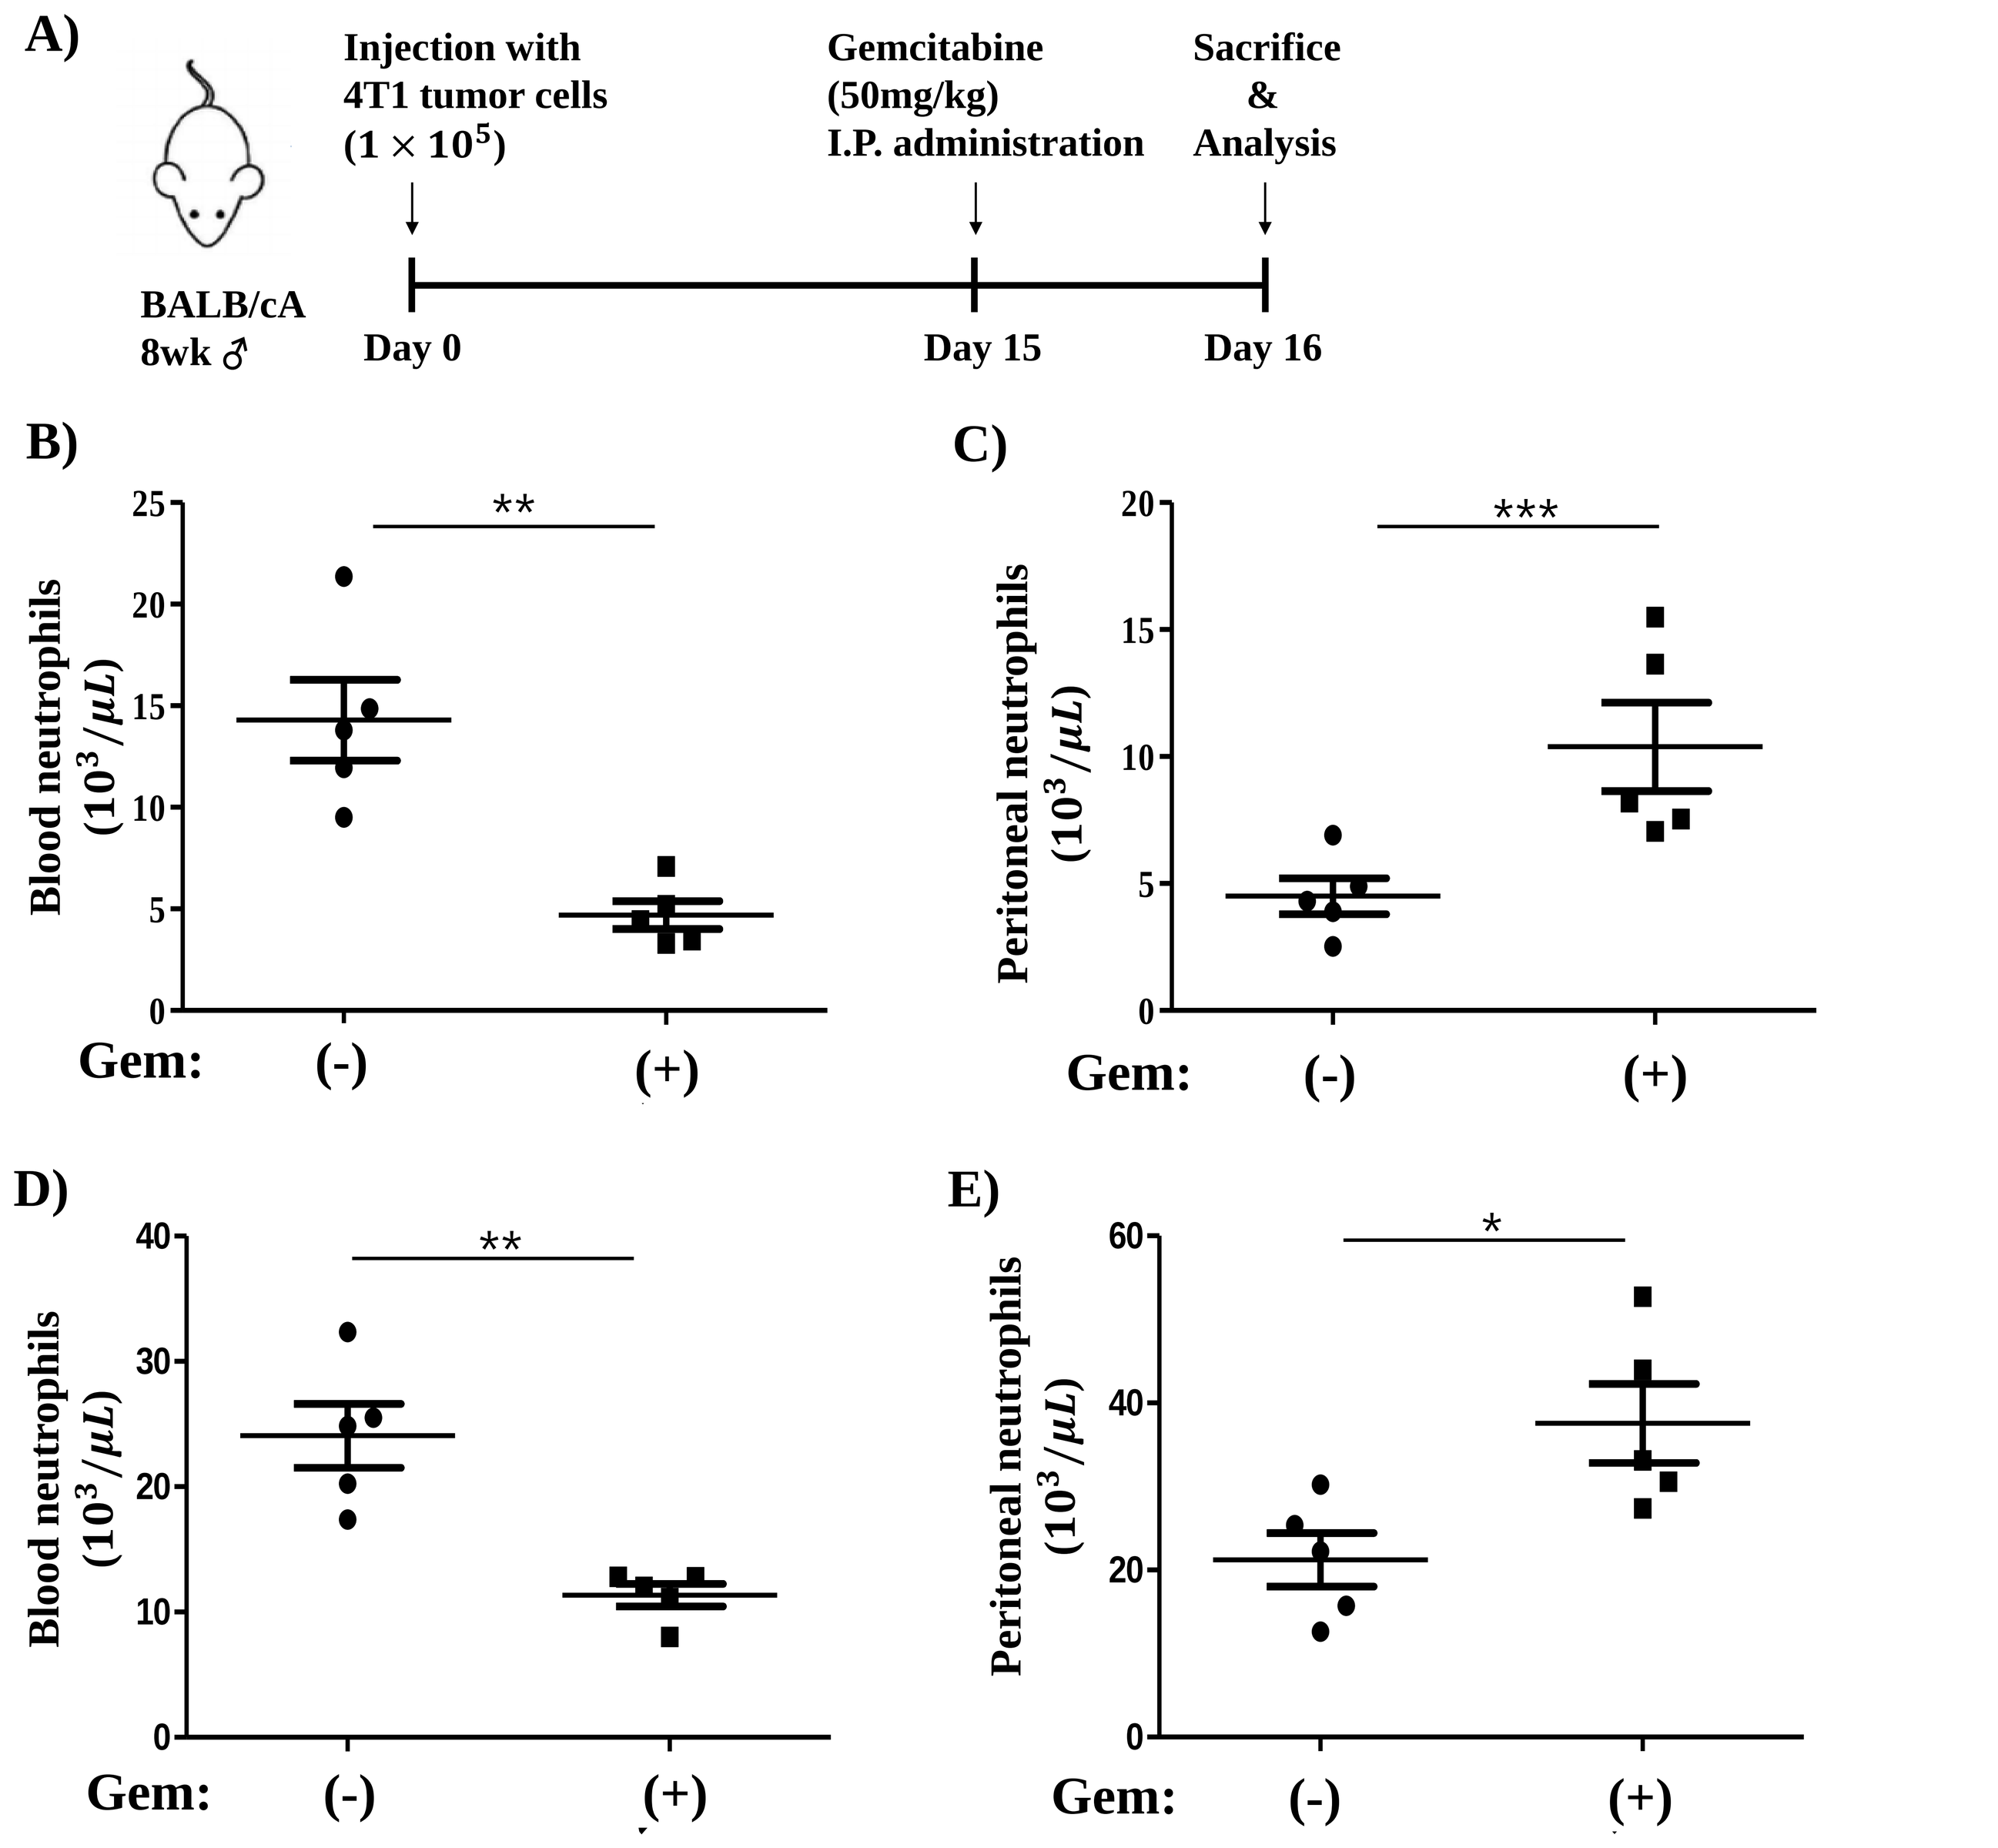
A)
Injection with 4T1 tumor cells (1 × 10^5)
Gemcitabine (50mg/kg) I.P. administration
Sacrifice & Analysis
BALB/cA 8wk ♂
Day 0
Day 15
Day 16
B)
Blood neutrophils (10^3/μL)
C)
Peritoneal neutrophils (10^3/μL)
D)
Blood neutrophils (10^3/μL)
E)
Peritoneal neutrophils (10^3/μL)
Gem: (-) (+)

A) A diagram of experimental design. **B-E)** the number of neutrophils and WBCs was determined by CBC analysis. Gemcitabine was administered by intraperitoneal injection (i.p.) in a dose of 50 mg/kg for 24h. Each group contains five mice. The bars represent the mean ± SD. Student's t-test was performed to determine the p values, and p values less than 0.05 were considered statistically significant. *p<0.05, **p<0.01, ***p<0.005.

2. Gemcitabine induces expression of neutrophil attracting chemokines and receptors in the peritoneal cells and the tumor tissues.

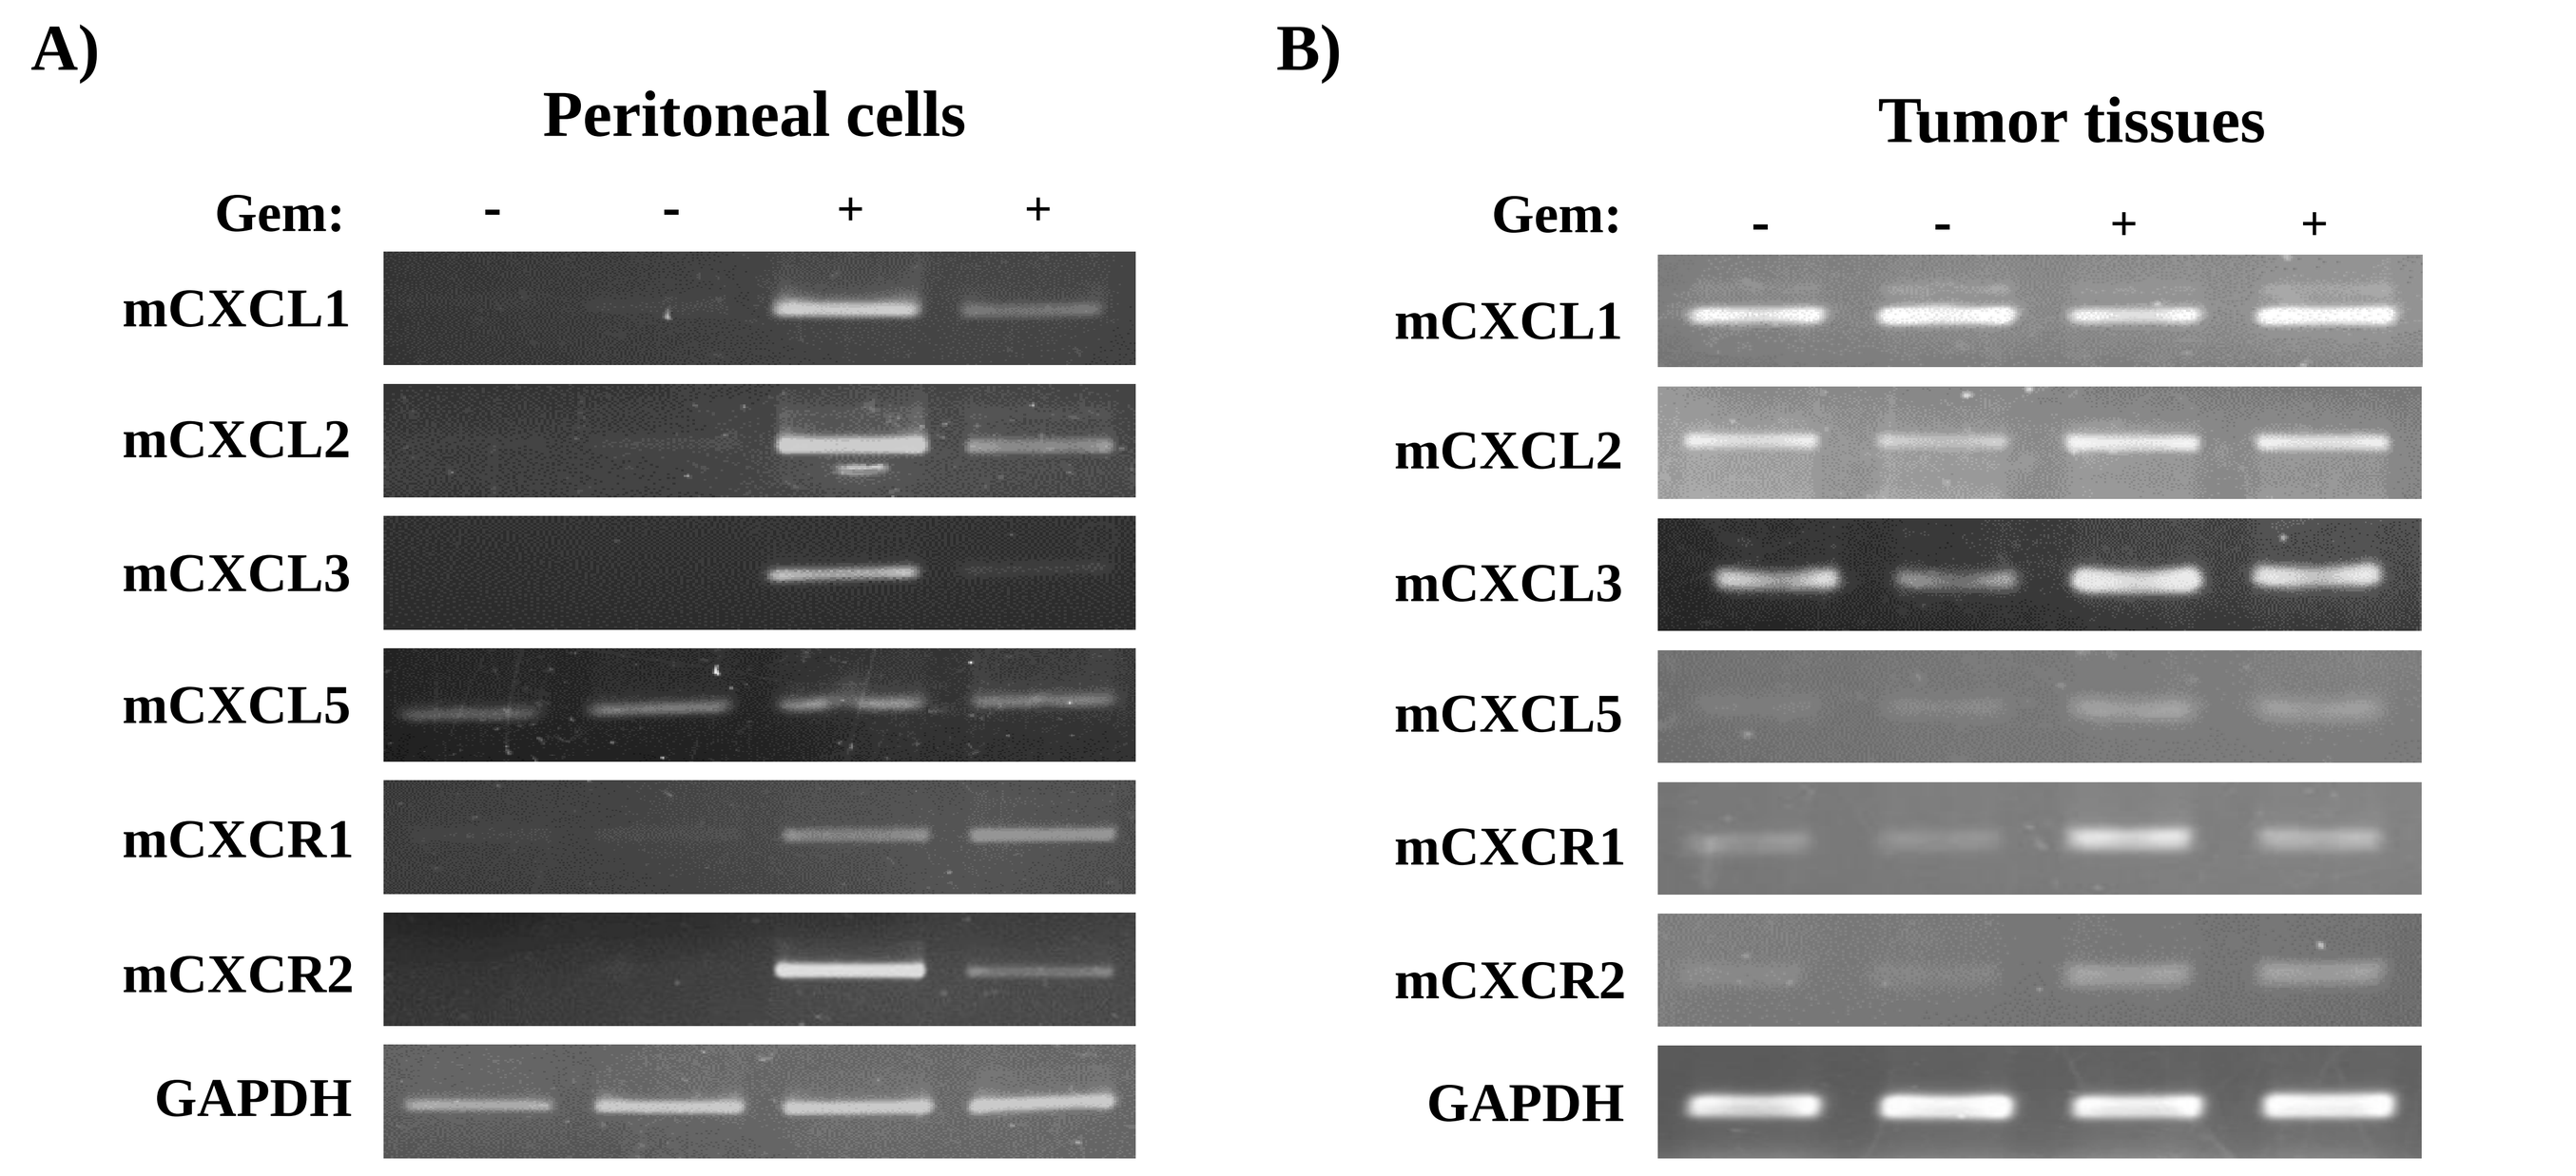
A)
Peritoneal cells
B)
Tumor tissues
Gem: - - + +
mCXCL1
mCXCL2
mCXCL3
mCXCL5
mCXCR1
mCXCR2
GAPDH

A and B) The mRNA levels of chemokines and its receptors from peritoneal cells and tumor tissues were analyzed by conventional RT-PCR. The mRNA expression of chemokines and receptors known for attracting neutrophil chemotaxis was elevated in response to gemcitabine treatment.

3. Reactive oxygen species (ROS) is the major cause that induces neutrophil transmigration and chemokine expressions in gemcitabine-induced neutropenia.

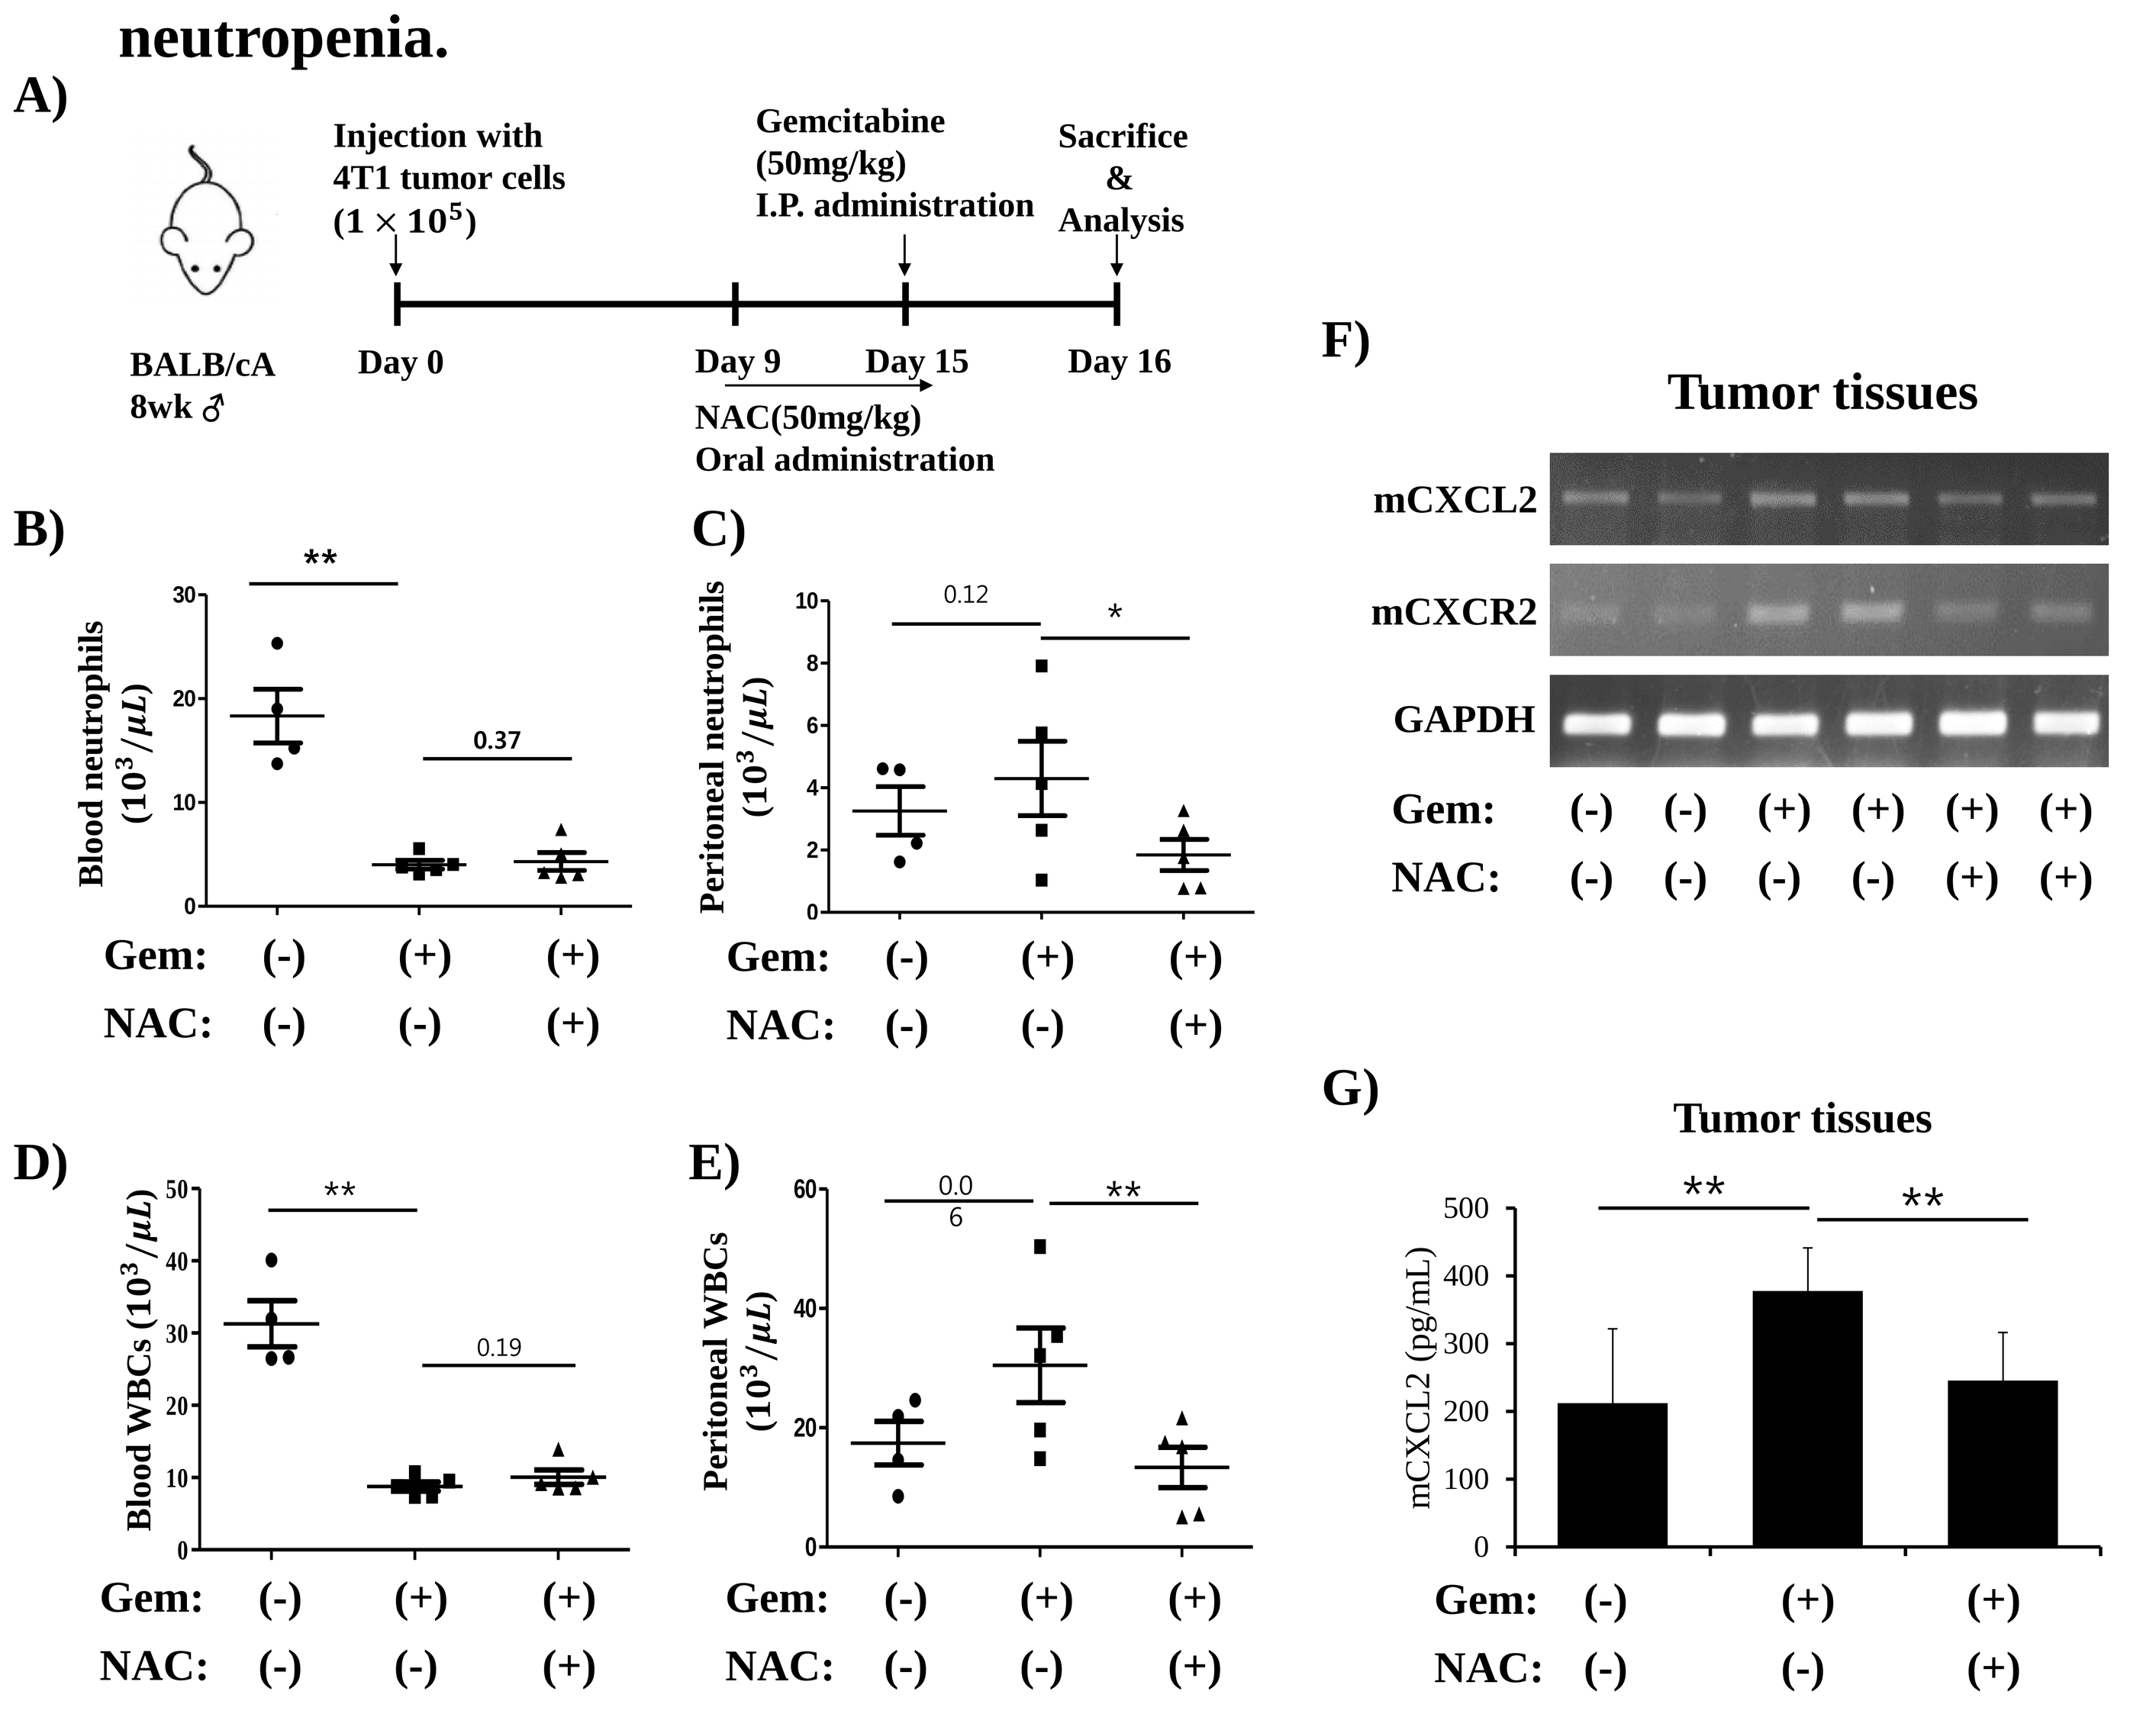
A)
Injection with 4T1 tumor cells (1 × 10^5)
Gemcitabine (50mg/kg) I.P. administration
Sacrifice & Analysis
BALB/cA 8wk ♂
Day 0
Day 9
Day 15
Day 16
NAC(50mg/kg) Oral administration
B)
Blood neutrophils (10^3/μL)
C)
Peritoneal neutrophils (10^3/μL)
D)
Blood WBCs (10^3/μL)
E)
Peritoneal WBCs (10^3/μL)
F)
Tumor tissues
mCXCL2
mCXCR2
GAPDH
G)
Tumor tissues
mCXCL2 (pg/mL)
Gem:
NAC:

A) A diagram of experimental design. Each group contains five mice. Gemcitabine was administered intraperitoneally at a dose of 50 mg/kg. N-acetyl-L-cysteine (NAC) was orally administered at a dose of 250 mg/kg daily for 6 consecutive days. The number of **B,C)** neutrophils and **D,E)** WBCs in blood and peritoneum was determined by CBC analysis. **F)** the mRNA levels of mCXCL2 and mCXCR2 from tumor tissues was analyzed by conventional RT-PCR. **G)** the protein level of mCXCL2 from tumor tissues was analyzed by ELISA. The bars represent the mean ± SD. Student's t-test was performed to determine the p values, and p values less than 0.05 were considered statistically significant. *p<0.05, **p<0.01, ***p<0.005.

4. Reparixin, a CXCR2 antagonist, inhibits gemcitabine-induced neutropenia.

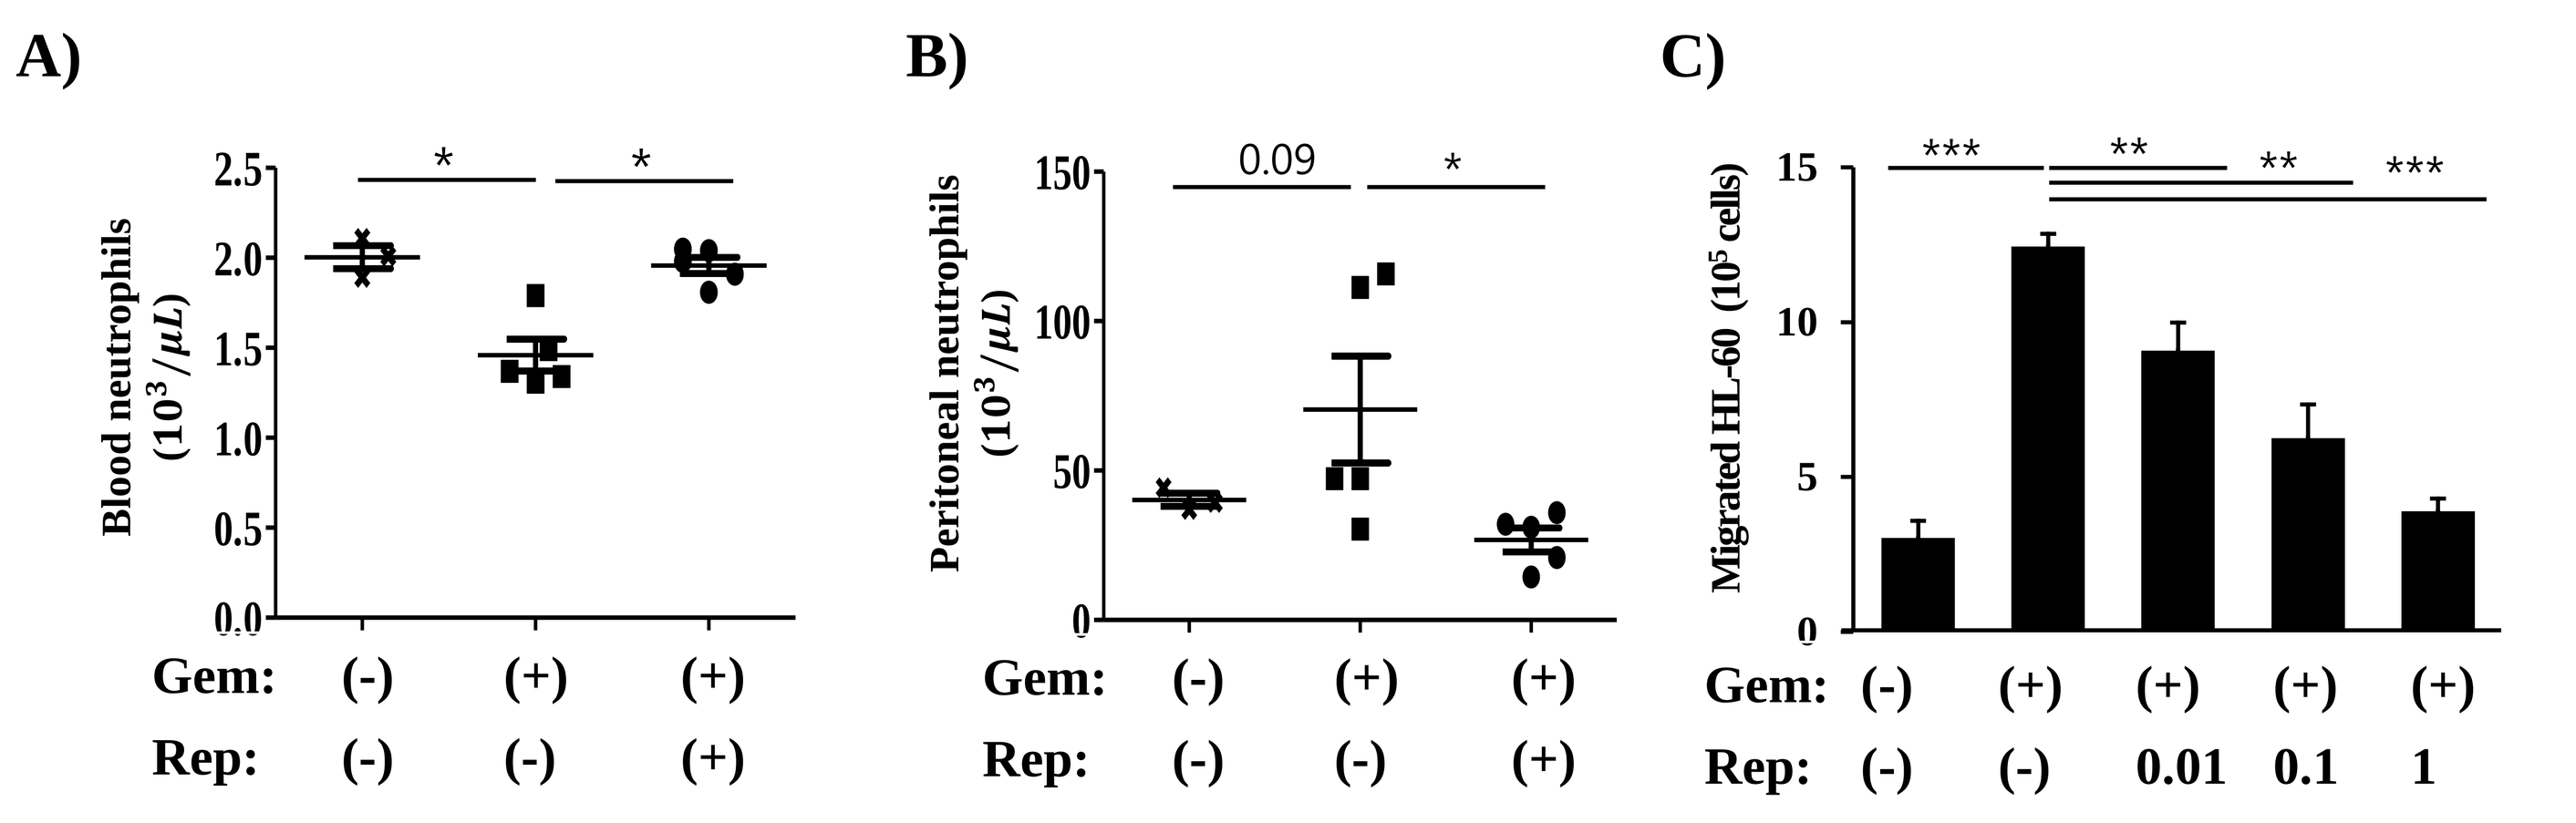
A)
Blood neutrophils (10^3/μL)
B)
Peritoneal neutrophils (10^3/μL)
C)
Migrated HL-60 (10^5 cells)
Gem:
Rep:
0.01
0.1
1

(A, B) Gemcitabine was administered by intraperitoneal injection (i.p.) at a dose of 50 mg/kg. Reparixin, a CXCR2 antagonist, was administered by i.p. injection at a dose of 30mg/kg and after 24h, number of neutrophil in blood **(A)** and peritoneum **(B)** was determined by CBC analysis. **(C)** The supernatant of gemcitabine treated THP-1 cells was placed in the bottom chamber. And HL-60 cells were seeded in the upper chamber and treated with various concentrations of CXCL8 antagonist, SB 225002 (0.01, 0.1 or 1 mM). Migrated HL-60 cell was counted after 24hr incubation in assay chamber. The bars represent the mean ± SD. Student's t-test was performed to determine the p values, and p values less than 0.05 were considered statistically significant. *p<0.05, **p<0.01, ***p<0.005.

4. Gemcitabine induces the phosphorylation of PKCδ and ERK1/2 MAPK.

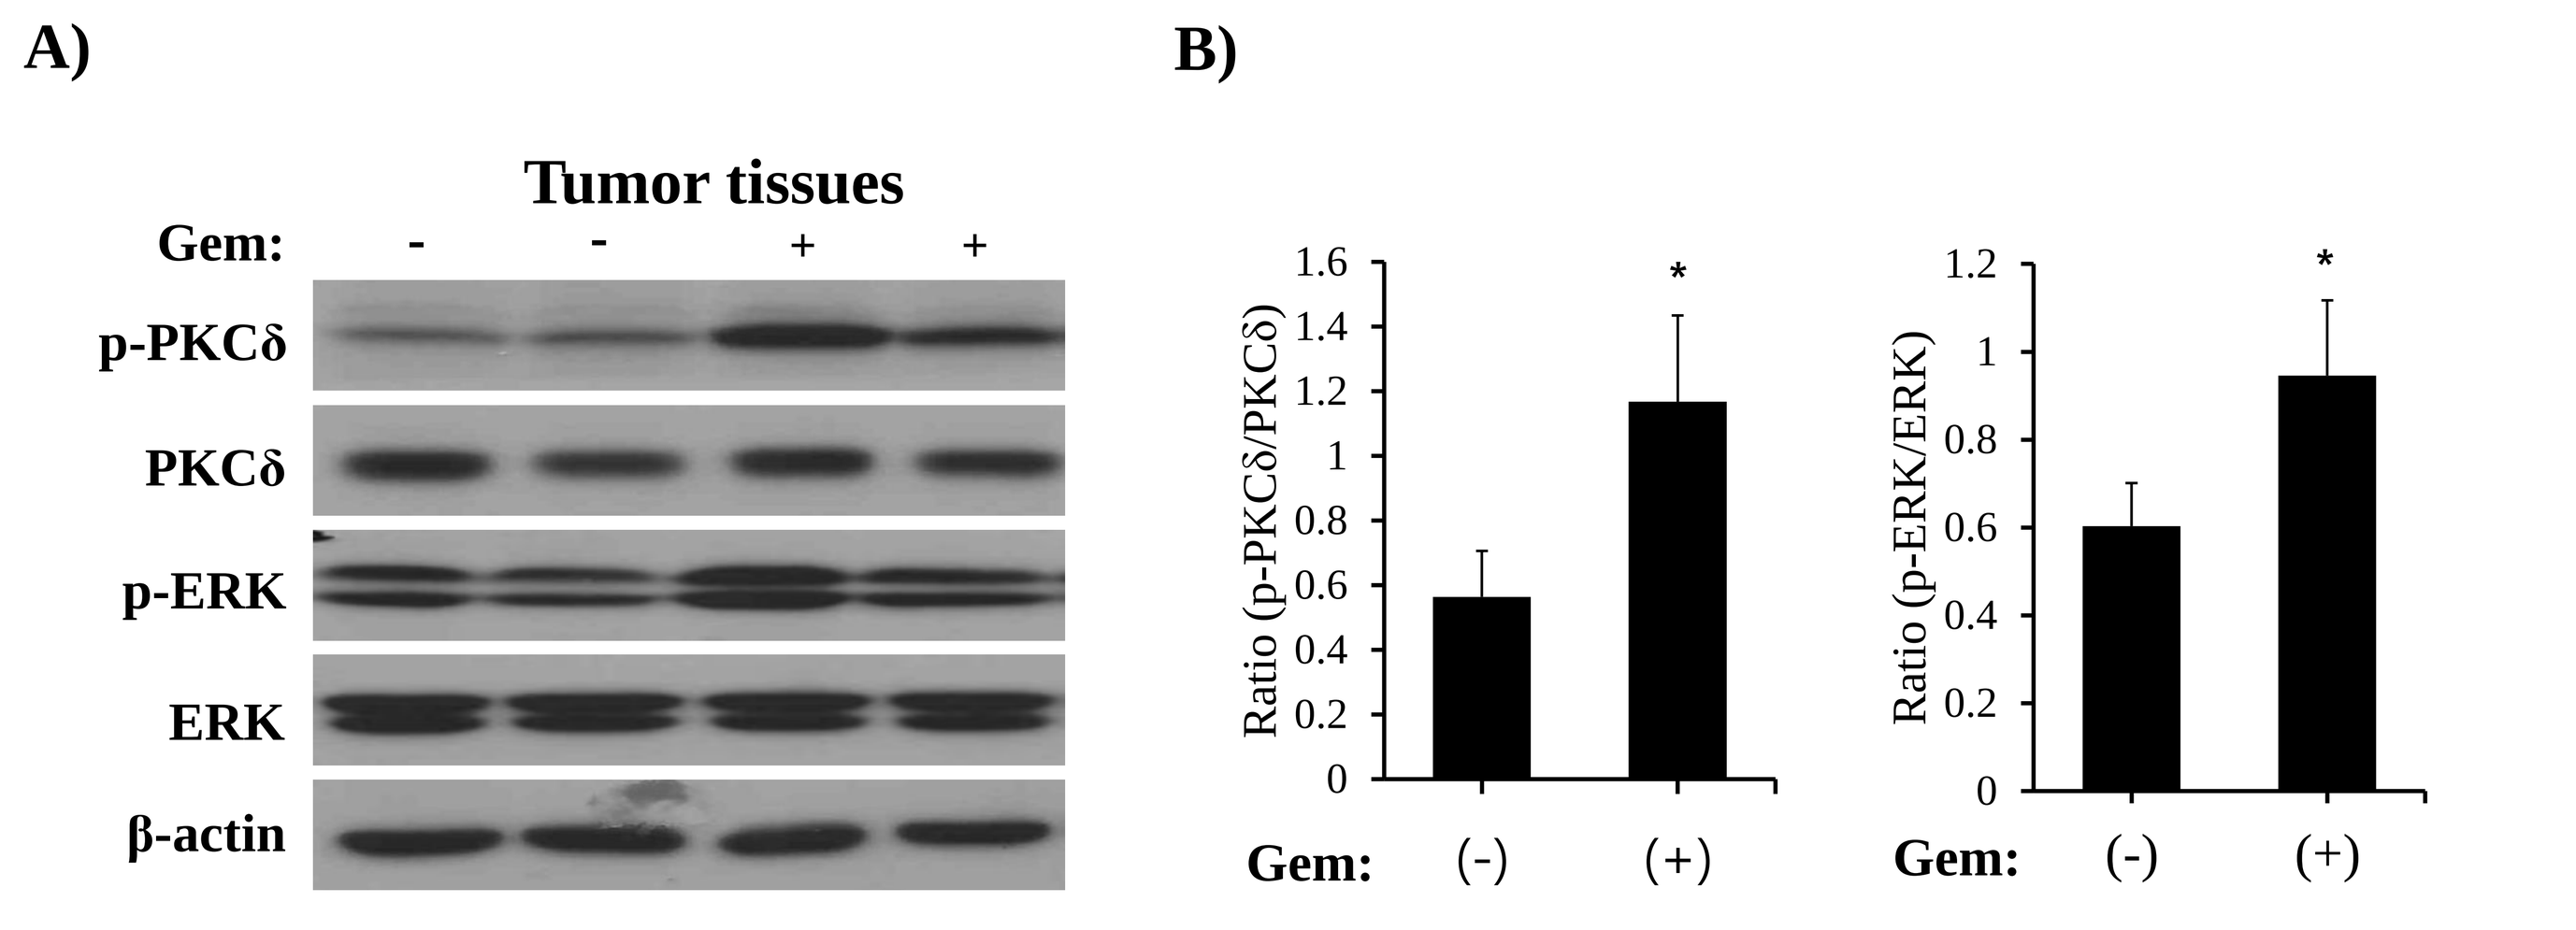
A)
Tumor tissues
Gem: - - + +
p-PKCδ
PKCδ
p-ERK
ERK
β-actin
B)
Ratio (p-PKCδ/PKCδ)
Ratio (p-ERK/ERK)
Gem: (-) (+)

A) Extracts from gemcitabine treated tumor tissues were analyzed by Western blot. Gemcitabine increased the phosphorylation of PKCδ and ERK1/2 compared to the control. **B)** Data are presented as mean fold increases (±SD) in treated groups over basal values from three independent experiments. Student's t-test was performed to determine the p values, and p values less than 0.05 were considered statistically significant. *p<0.05, **p<0.01, ***p<0.005.

5. Gemcitabine exerts synergistic effects with conditioned medium of MBA-MD-231 cells on ERK1/2 MAPK activation and CXCL8 production *in vitro*.

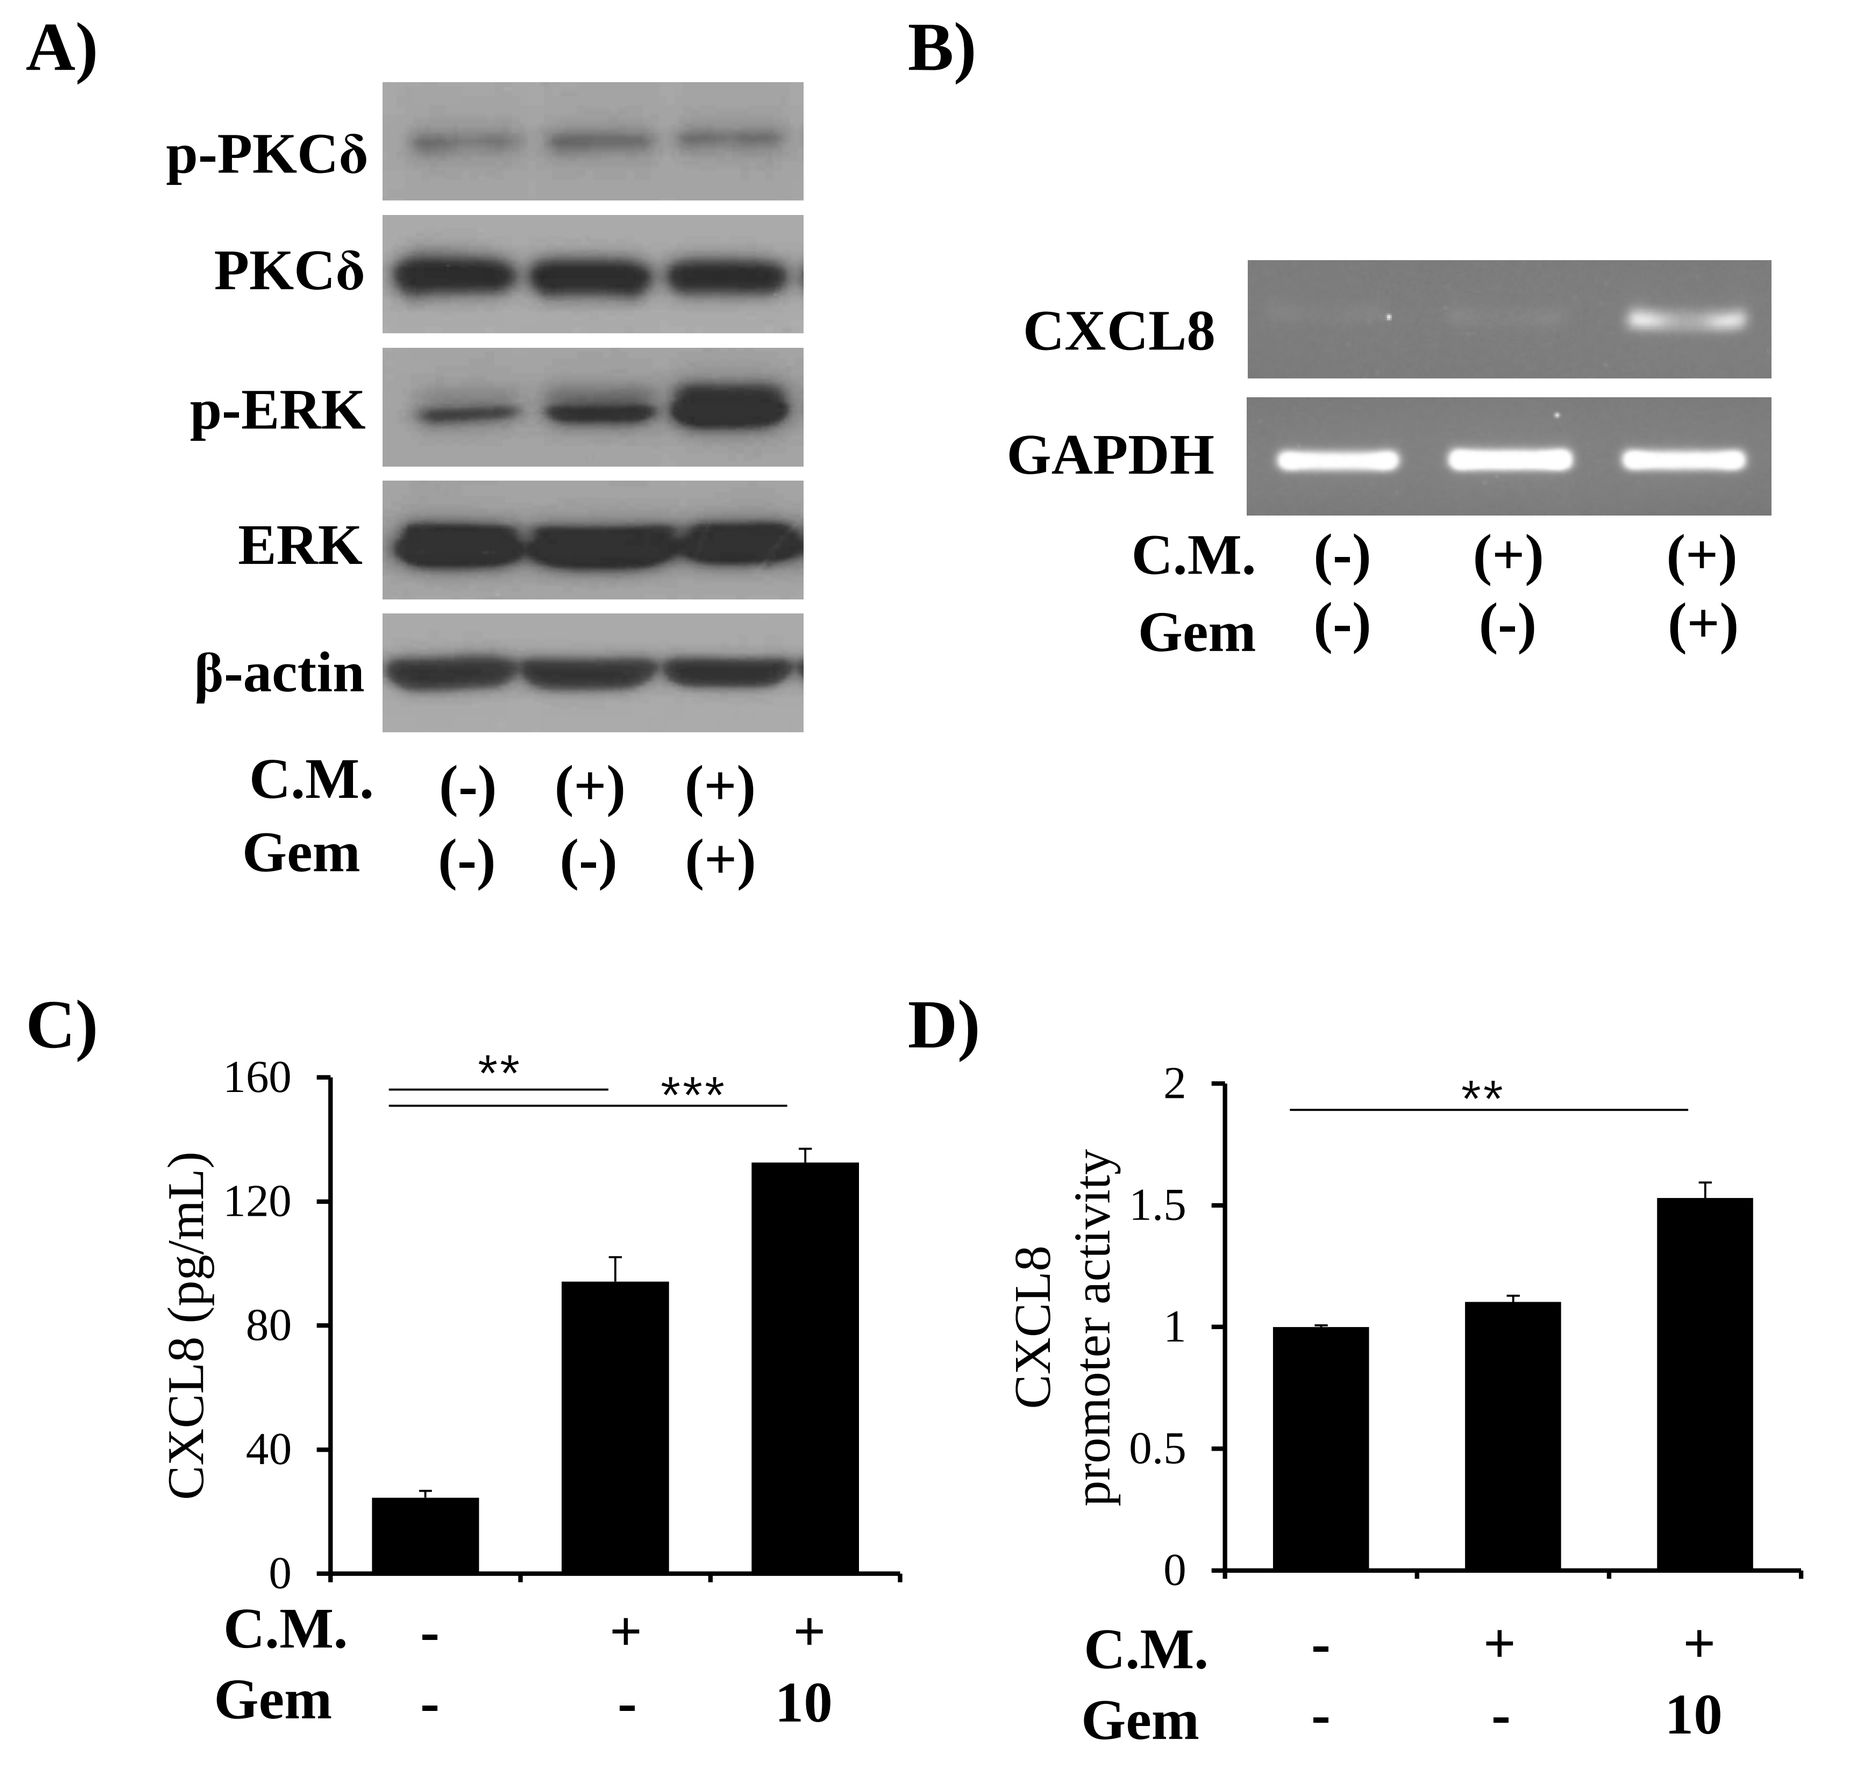
A)
p-PKCδ
PKCδ
p-ERK
ERK
β-actin
B)
CXCL8
GAPDH
C.M.
Gem
C)
CXCL8 (pg/mL)
D)
CXCL8 promoter activity

THP-1 cells were co-treated with MDA-MB-231 conditioned medium (C.M.) and 10μg/mL of gemcitabine. **A)** Cellular extracts were analyzed by western blot. **B)** The mRNA level of CXCL8 was analyzed by conventional RT-PCR. **C)** The protein concentration of CXCL8 from THP-1 supernatants was determined by ELISA. **D)** Reporter construct containing luciferase gene regulated by CXCL8 promoter sites was transfected into A549 cells, and the expression of the luciferase gene was analyzed by Dual-Glo luciferase system. Student's t-test was performed to determine the p values, and p values less than 0.05 were considered statistically significant. *p<0.05, **p<0.01, ***p<0.005.

Conclusion

- Circulating neutrophils undergo extravasation in response to a chemotherapeutic agent, gemcitabine, by increasing their chemotactic activity.
- ROS generation and chemokine-CXCR2 interaction play key roles in gemcitabine-induced neutrophil transmigration as confirmed by treatment of NAC (antioxidant) and reparixin (CXCR2 antagonist).
- THP-1 cells exhibit slightly elevated PKCδ activity when cultured in the conditioned medium of breast cancer cells and produce CXCL8 synergistically with gemcitabine treatment by upregulating ERK1/2 MAPK activity.
- We suggest that it is critical to control PKCδ activity in cancer patients because PKCδ-activated intra-tumoral macrophages produce neutrophil attracting chemokines synergistically with chemotherapeutic agents, which induce CIN.

References

[1] Crawford, J., Dale, D. C., & Lyman, G. H. (2004). **Cancer,** 100(2), 228-237.

[2] Baggiolini, M., Walz, A., & Kunkel, S. L. (1989). **Journal of Clinical Investigation**, 84(4), 1045.

[3] Coffelt, S. B., Wellenstein, M. D., & de Visser, K. E. (2016). **Nature Reviews Cancer**, *16*(7), 431-446.

[4] Plunkett, W., et al. (1995, August). **In Seminars in oncology** (Vol. 22, No. 4 Suppl 11, pp. 3-10).

[5] Fiorini, C., et al. (2015). **Biochimica et Biophysica Acta (BBA)-Molecular Cell Research**, *1853*(3), 549-560.